Phase 1 Results with Anti-CD19 Allogeneic CAR T ALLO-501/501A in Relapsed/Refractory Large B-Cell Lymphoma (r/r LBCL)

Locke FL[1], Lekakis LJ[2], Eradat HA[3], Munoz J[4], Tees M[5], de Vos S[6], Nath R[7], Stevens DA[8], Malik SA[9], Popplewell LL[10], Hamadani M[11], Oluwole OO[12], Perales M-A[13], Miklos DB[14], Fisher PW[15], Goyal L[15], Kaufman GP[15], Kai K[15], Balakumaran A[15], Neelapu SS[16]

[1]Moffitt Cancer Center, Tampa, FL; [2]University of Miami, Miami, FL; [3]Ronald Reagan UCLA Medical Center, Los Angeles, CA; [4]Mayo Clinic Comprehensive Cancer Center, Phoenix, AZ; [5]Colorado Blood Cancer Institute, Denver, CO; [6]UCLA, Los Angeles, CA; [7]Banner Health Center, Gilbert, AZ; [8]Norton Cancer Institute, Louisville, KY; [9]Sarah Cannon Transplant & Cellular Therapy Program at St. David's South Austin Medical Center, Austin, TX; [10]City of Hope National Medical Center, Duarte, CA; [11]Medical College of Wisconsin, Milwaukee, WI; [12]Vanderbilt University Medical Center, Nashville, TN; [13]Memorial Sloan Kettering Cancer Center, New York, NY; [14]Stanford University, Stanford, CA; [15]Allogene Therapeutics, South San Francisco, CA; [16]The University of Texas MD Anderson Cancer Center; Houston, TX.

Abstract ID: 2517

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.

Background

- Autologous CAR T cell therapies have transformed the treatment of relapsed/refractory non-Hodgkin lymphoma (r/r NHL) but, due to a lengthy and cumbersome manufacturing processes, are not available to all eligible patients (pts).
- ALLO-501A is an HLA-unmatched, off-the-shelf, investigational, anti-CD19 allogeneic CAR T cell product administered as a one-time treatment that is potentially capable of inducing durable remissions in r/r LBCL pts. ALLO-501 is similar to ALLO-501A, except for the inclusion of a rituximab off switch.
- Two studies, ALPHA (NCT03939026) and ALPHA2 (NCT04416984), were undertaken to evaluate ALLO-501 and ALLO-501A in pts with r/r NHL.
- This update focuses on a subgroup of pts from the ALPHA and ALPHA2 studies who were treated with the regimen currently being evaluated in ALPHA2, a potentially pivotal Phase 2 trial in pts with r/r LBCL.

Figure 1. ALLO-501/501A

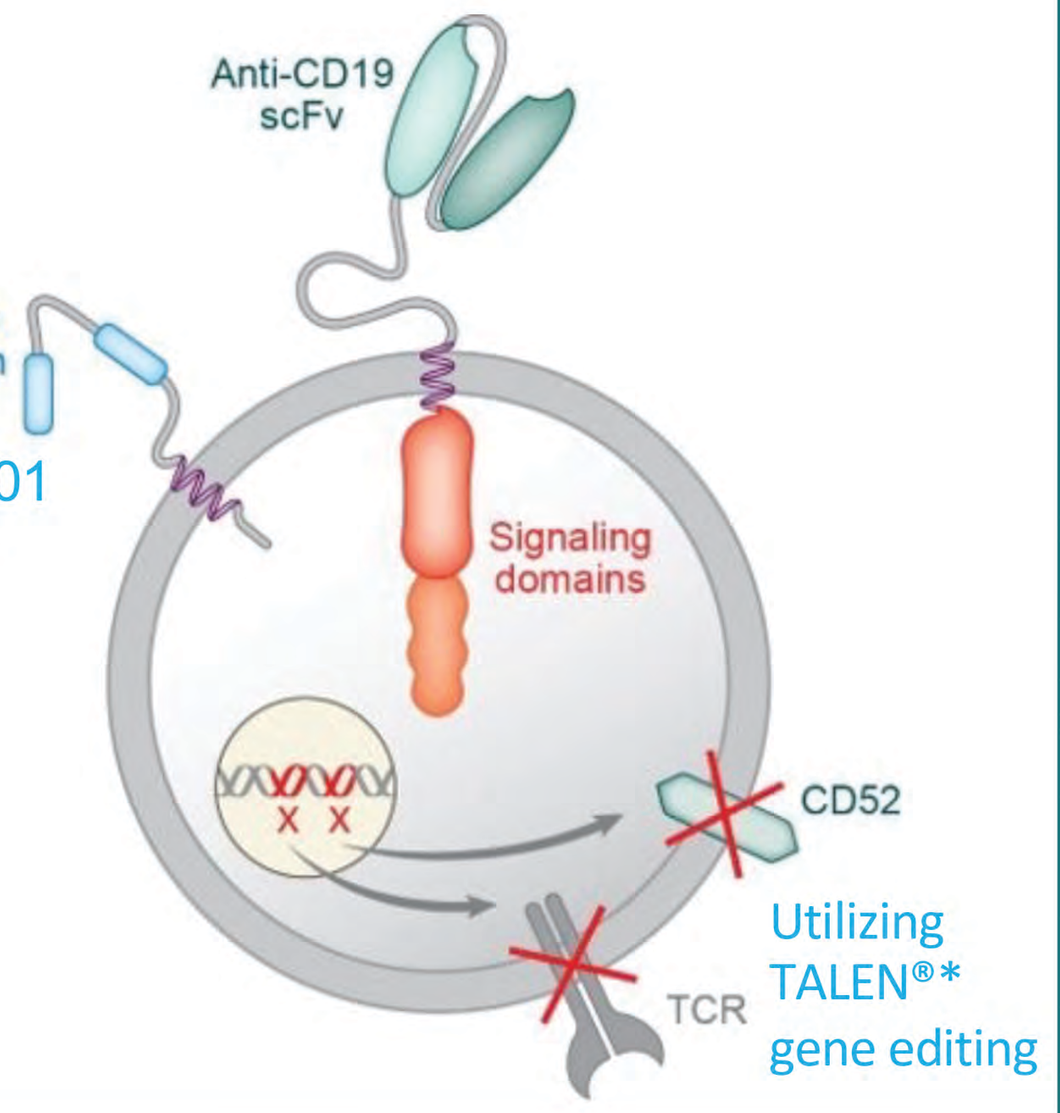

Anti-CD19 scFv
Rituximab recognition domains
ALLO-501 only
Signaling domains
CD52
TCR
Utilizing TALEN®* gene editing

Methods

Subgroup Selection and Analysis Sets

- CAR T-naïve pts with r/r LBCL (N=33) were treated with escalating doses of ALLO-501/501A manufactured with the Phase 2 process after lymphodepletion with FC and varying doses of ALLO-647. This subgroup comprises the safety set.
- Based on the overall Phase 1 experience, lymphodepletion with FCA90 and a single infusion of ALLO-501A was selected for evaluation in the Phase 2 portion of ALPHA2.
- 12 of the 33 pts in the safety set received lymphodepletion with FCA90, the selected Phase 2 lymphodepletion regimen. These patients comprise the efficacy and translational analysis set.

Endpoints

- Safety, tolerability, efficacy (overall response rate [ORR], complete response [CR] rate, and duration of response [DOR])
- T cell kinetics as measured by transgene levels in the peripheral blood
- Leukocyte recovery

Figure 2. Dosing & Administration With Selected Phase 2 Treatment Regimen

3-day lymphodepletion with FCA90	Single-dose of ALLO-501A or ALLO-501	Follow-up (ongoing)
F: fludarabine 30 mg/m²/day **C:** cyclophosphamide 300 mg/m²/day **A:** ALLO-647 30 mg/day (total dose: 90 mg)	120-360 x 10⁶ viable CAR+ cells	

*TALEN® gene editing is a technology pioneered and controlled by Cellectis.

Baseline Patient and Disease Characteristics

- Pts were heavily pretreated and had unfavorable baseline disease characteristics.
- Two thirds of pts had stage IV disease and two thirds had an elevated LDH at time of study enrollment.
- 92% of pts had an ECOG Performance Status (PS) of 1.
- Half or more of the pts had an IPI score >2, extranodal disease, and/or prior hematopoietic cell transplant.
- One third of pts had double or triple hit lymphoma.

Table 1. Baseline Patient Characteristics

	Pts Treated with Phase 2 Regimen (N=12)
Age, median	60 years
Stage IV disease	67%
ECOG PS of 1	92%
Baseline LDH > ULN	67%
IPI score >2	50%
Germinal center subtype	50%
Double or triple hit	33%
Median # prior regimens	3
Prior transplant	50%
Extranodal disease	58%

ECOG = Eastern Cooperative Oncology Group; IPI = International Prognostic Index; IRR = infusion-related reactions; LDH = lactate dehydrogenase; ULN = upper limit of normal.

Treatment Experience and Follow Up

- All treated pts (100%) received study treatment as intended; each infused dose of allogeneic CAR T cells was manufactured and released per product specifications.
- 3-day median time from enrollment to initiation of study treatment.
- No patients required bridging therapy.
- The median follow-up was 32.9 months.

Safety and Tolerability

- No Gr ≥3 CRS events or any ICANS events were observed.
- No GvHD events were reported.
- Infections included low-grade viral reactivations detected on weekly protocol-required surveillance. Infections were manageable with routine treatment; no fatal infections were observed.
- Adverse events with FCA90 lymphodepletion were consistent with those in the full subgroup.

Table 2. Adverse Events of Interest

	All CAR T-Naïve r/r LBCL (N=33)		Pts Treated With Phase 2 Regimen (N=12)	
n (%)	All Gr	Gr ≥3	All Gr	Gr ≥3
CRS	8 (24)	0	4 (33)	0
ICANS	0	0	0	0
Neurotoxicity	13 (39)	2 (6)	4 (33)	0
GvHD	0	0	0	0
IRR	16 (49)	3 (9)	8 (67)	0
Infection	19 (58)	5 (15)	8 (67)	1 (8)
Prolonged Gr ≥3 Cytopenia	-	4 (12)	-	2 (17)

CRS = cytokine release syndrome; GvHD = graft-versus-host disease; ICANS = immune effector cell-associated neurotoxicity syndrome; IRR = infusion-related reactions.

All infections were reportable up to Month 3; Gr ≥3 infections were reportable through Month 60.

Efficacy

- The ORR was 67% and the CR rate was 58%, with a median duration of response of 23.1 months.

Table 3. Response Rates in Pts Treated With Selected Phase 2 Regimen

n (%)	Pts Treated with Phase 2 Regimen (N=12)
ORR	8 (67)
CR	7 (58)
6-month CR†	5 (42)

† Analysis of pts who had the opportunity to be followed through Month 6 or experienced disease progression prior to Month 6.

- 42% of pts sustained CR through Month 6; 80% of pts who were in CR at 6 months remain in CR.
- 3 pts remain in remission at 24+ months, with the longest remission ongoing beyond 31 months.

Figure 3. Swimmer Plot of Tumor Response in Patients Treated With Selected Phase 2 Regimen (N=12)

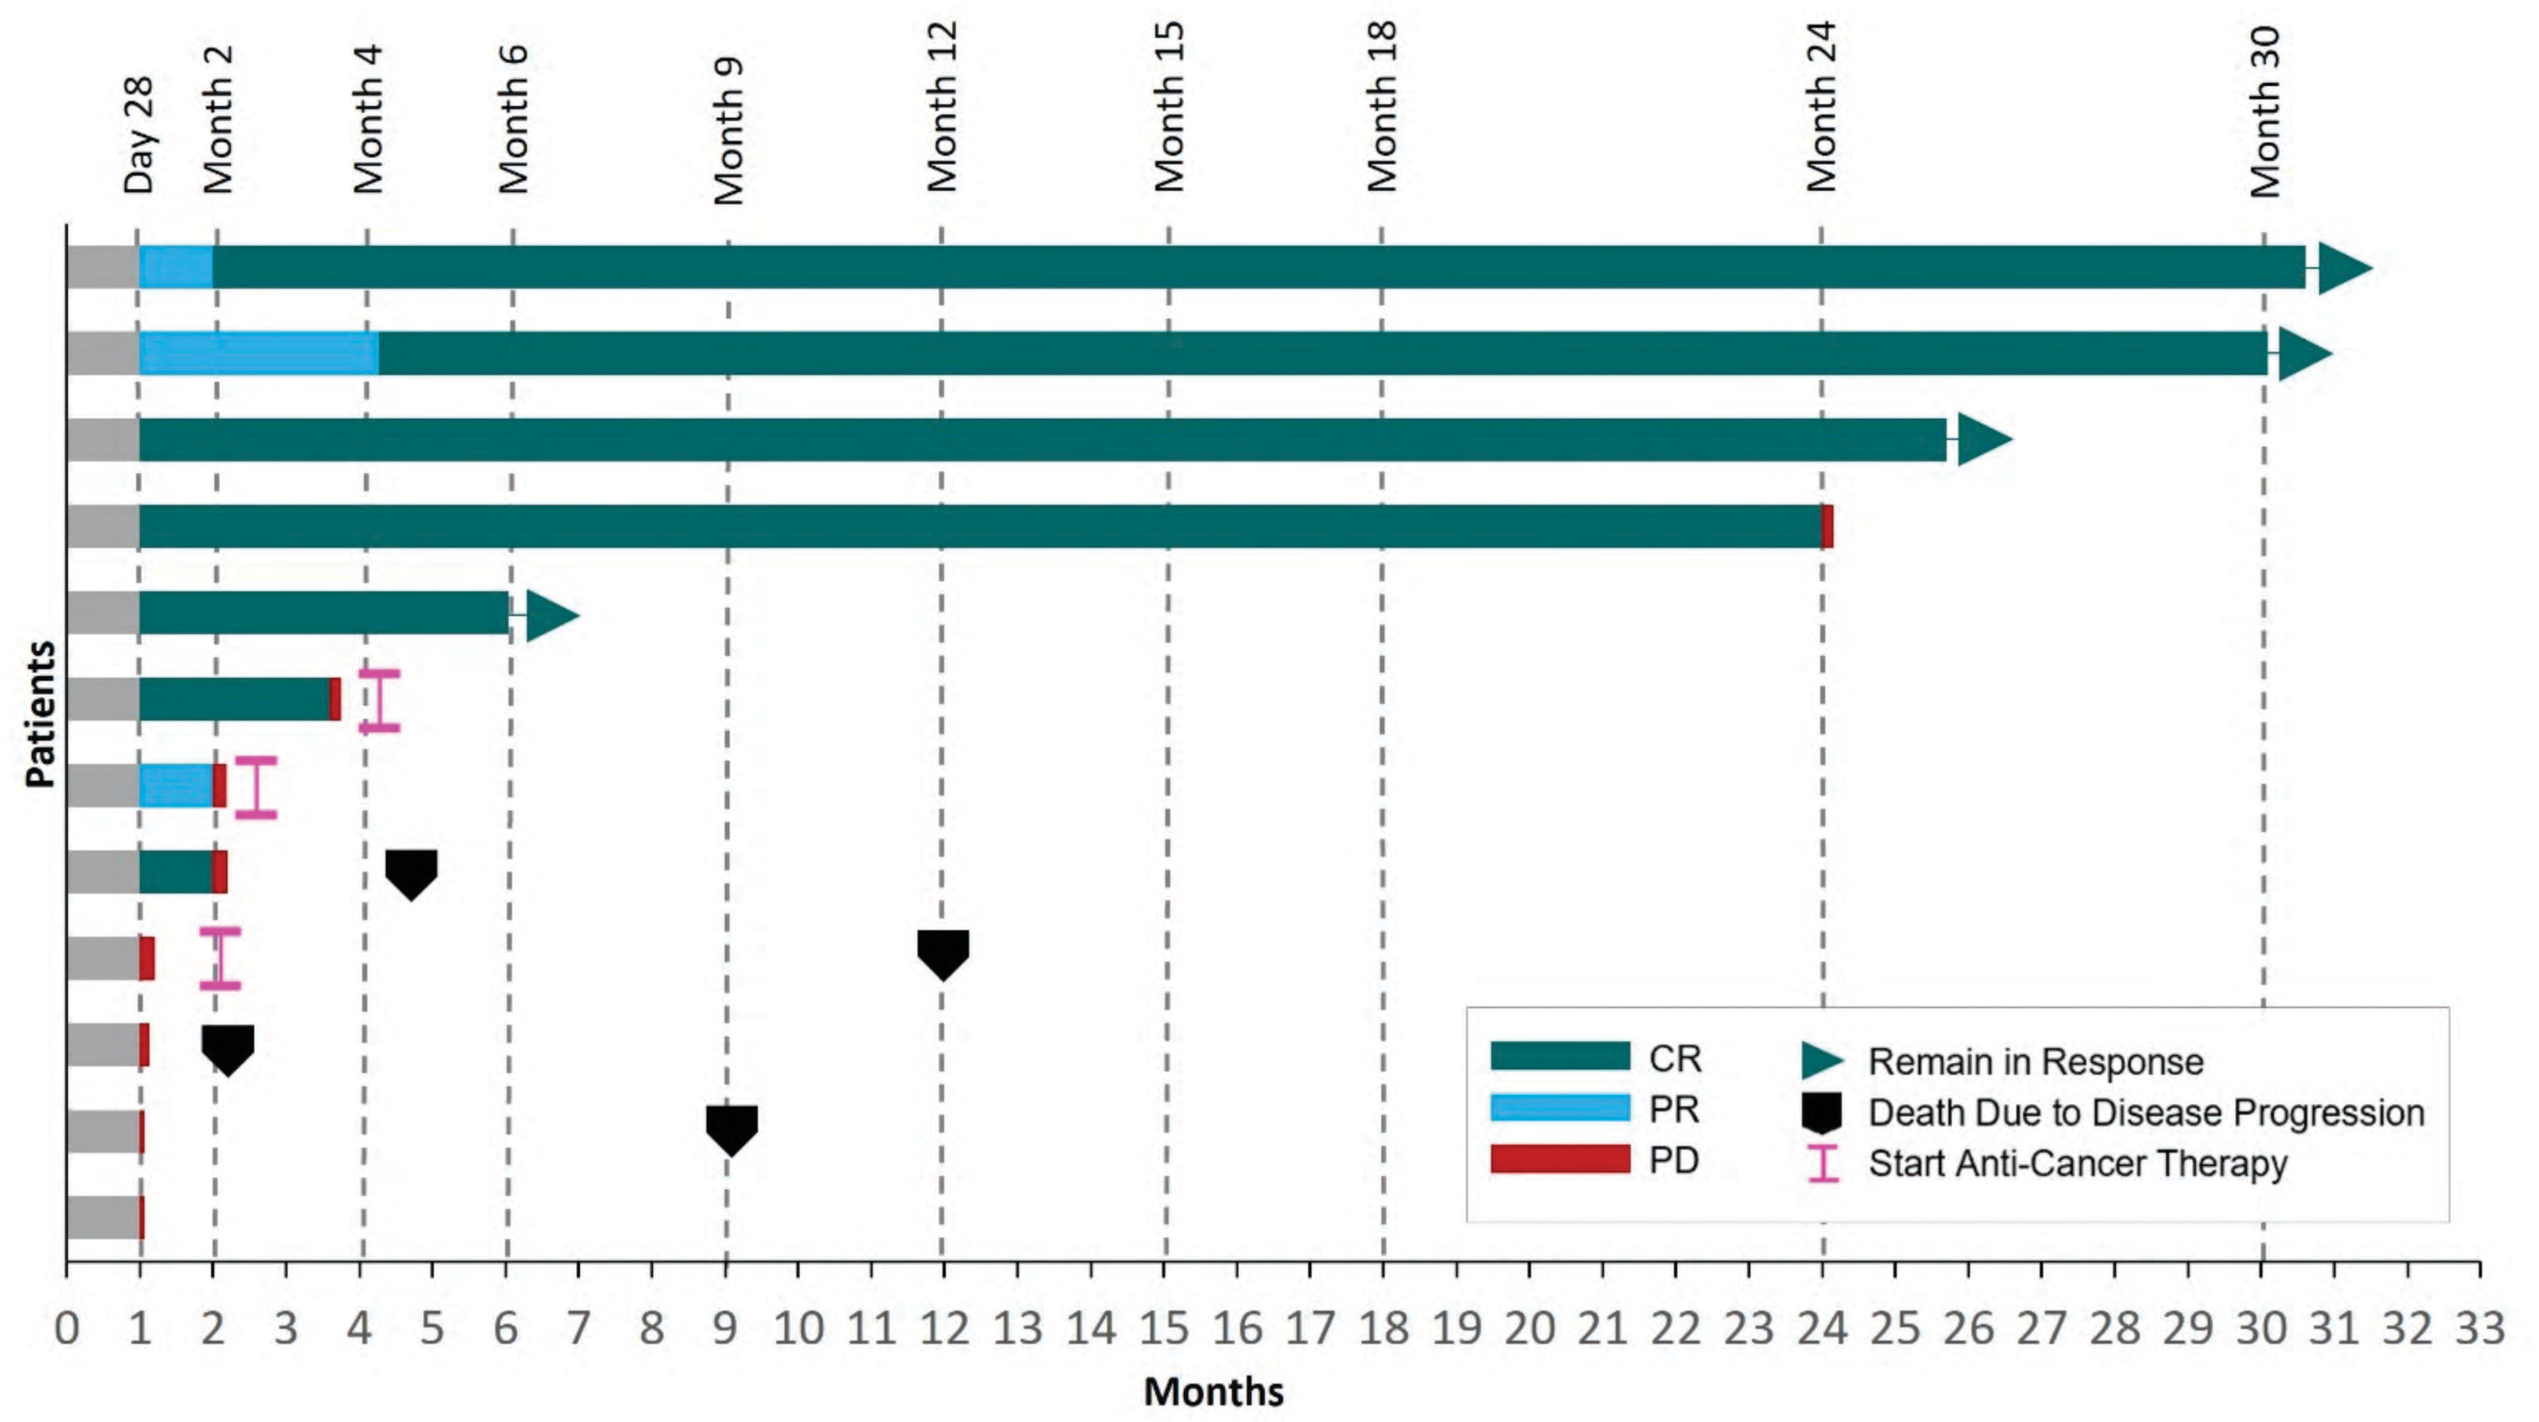

Day 28
Month 2
Month 4
Month 6
Month 9
Month 12
Month 15
Month 18
Month 24
Month 30
Patients
CR
PR
PD
Remain in Response
Death Due to Disease Progression
Start Anti-Cancer Therapy
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28 29 30 31 32 33
Months

Translational Analyses

CAR T Cell Expansion

- Calculated AUC (log of copies per microgram of DNA) per subject was markedly higher in peripheral blood sampled from Day 1 through Day 28 among responders vs non-responders.
- Allogeneic CAR T cell peak expansion and persistence was higher in responders vs non-responders (log of copies per microgram of DNA).

Figure 4. ALLO-501/ALLO-501A CAR T Cell Expansion is Associated With Response (N=11)‡

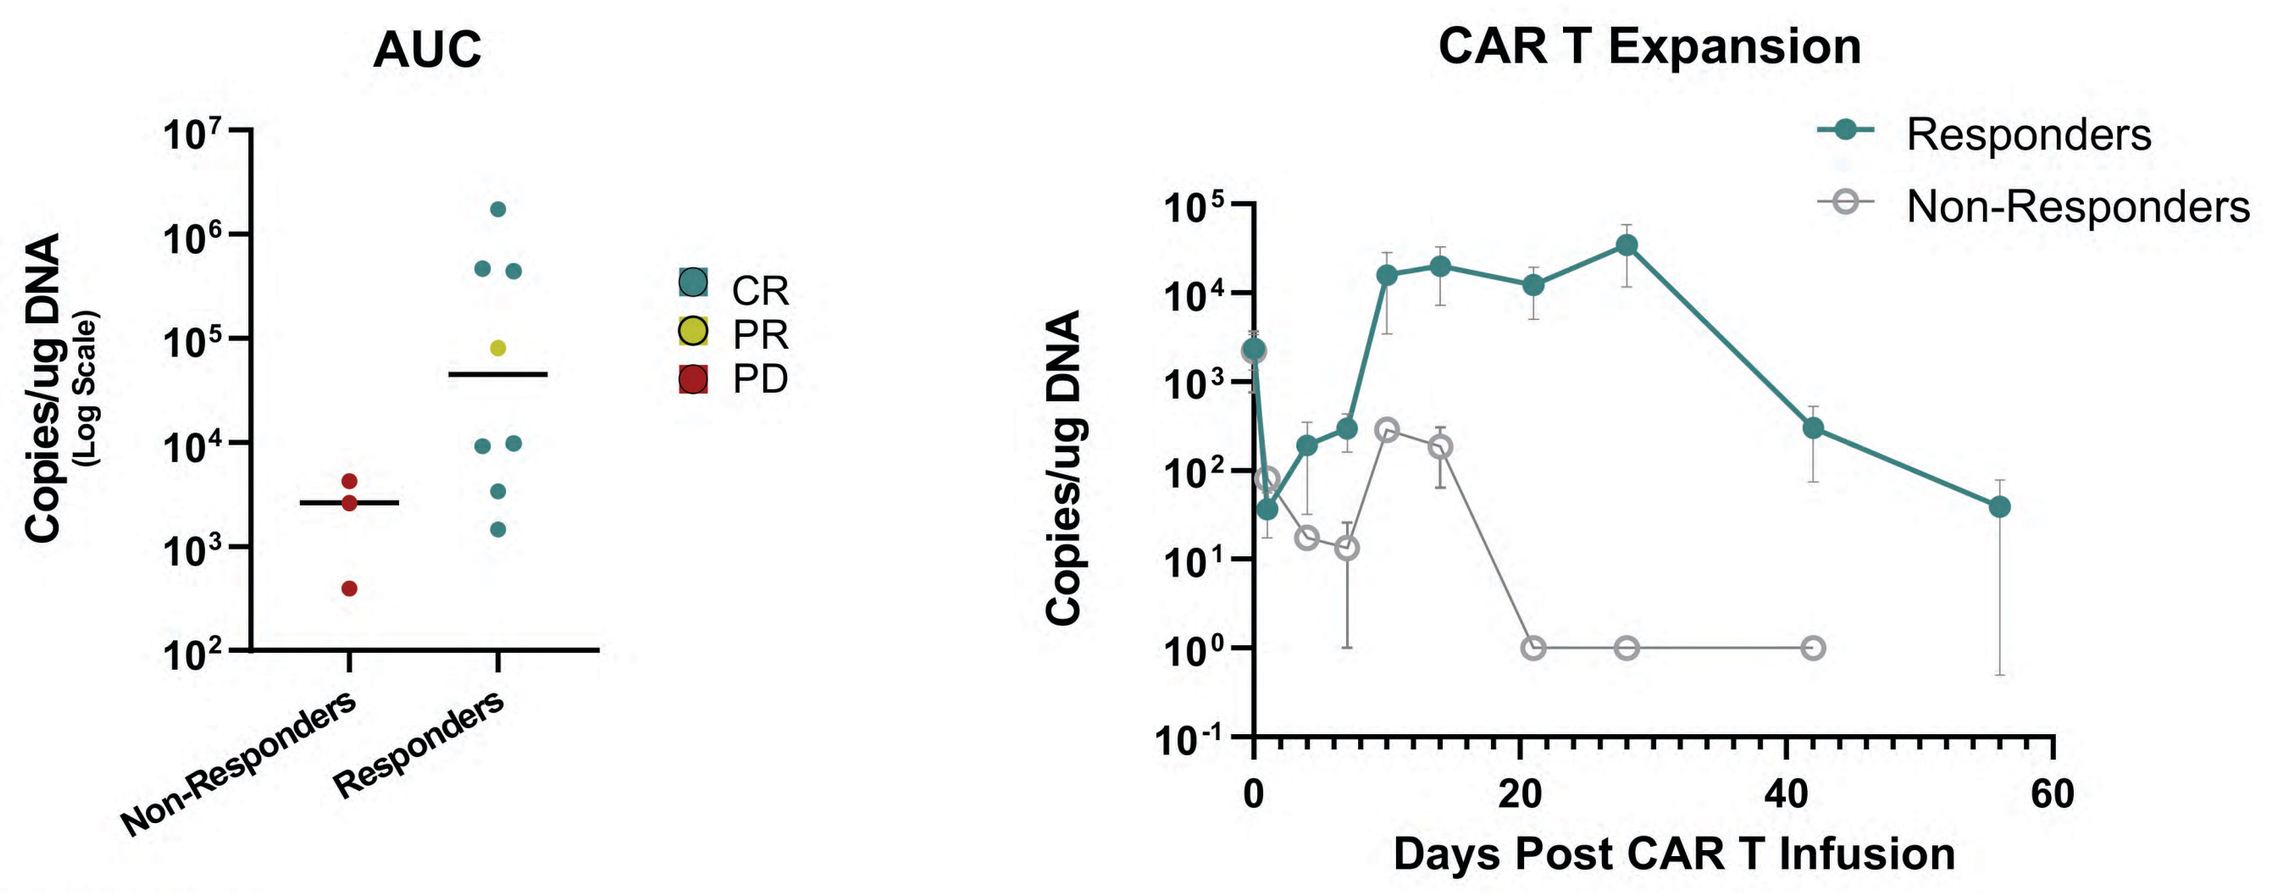

AUC
Copies/ug DNA (Log Scale)
Non-Responders
Responders
CR
PR
PD
CAR T Expansion
Responders
Non-Responders
Copies/ug DNA
Days Post CAR T Infusion

AUC: area under the curve.
‡ One subject did not have sample collection through Day 28 and was excluded from this analysis.

Translational Analyses, cont.

- Neutrophils recovered§ at a median of 7 days after ALLO-501/A.
- Lymphocytes recovered§ at a median of 21 days after ALLO-501/A.

§ Defined as Common Terminology Criteria for Adverse Events (CTCAE) v5.0: Grade <4.

Figure 5. Leukocyte Count Recovery With Selected Phase 2 Treatment Regimen

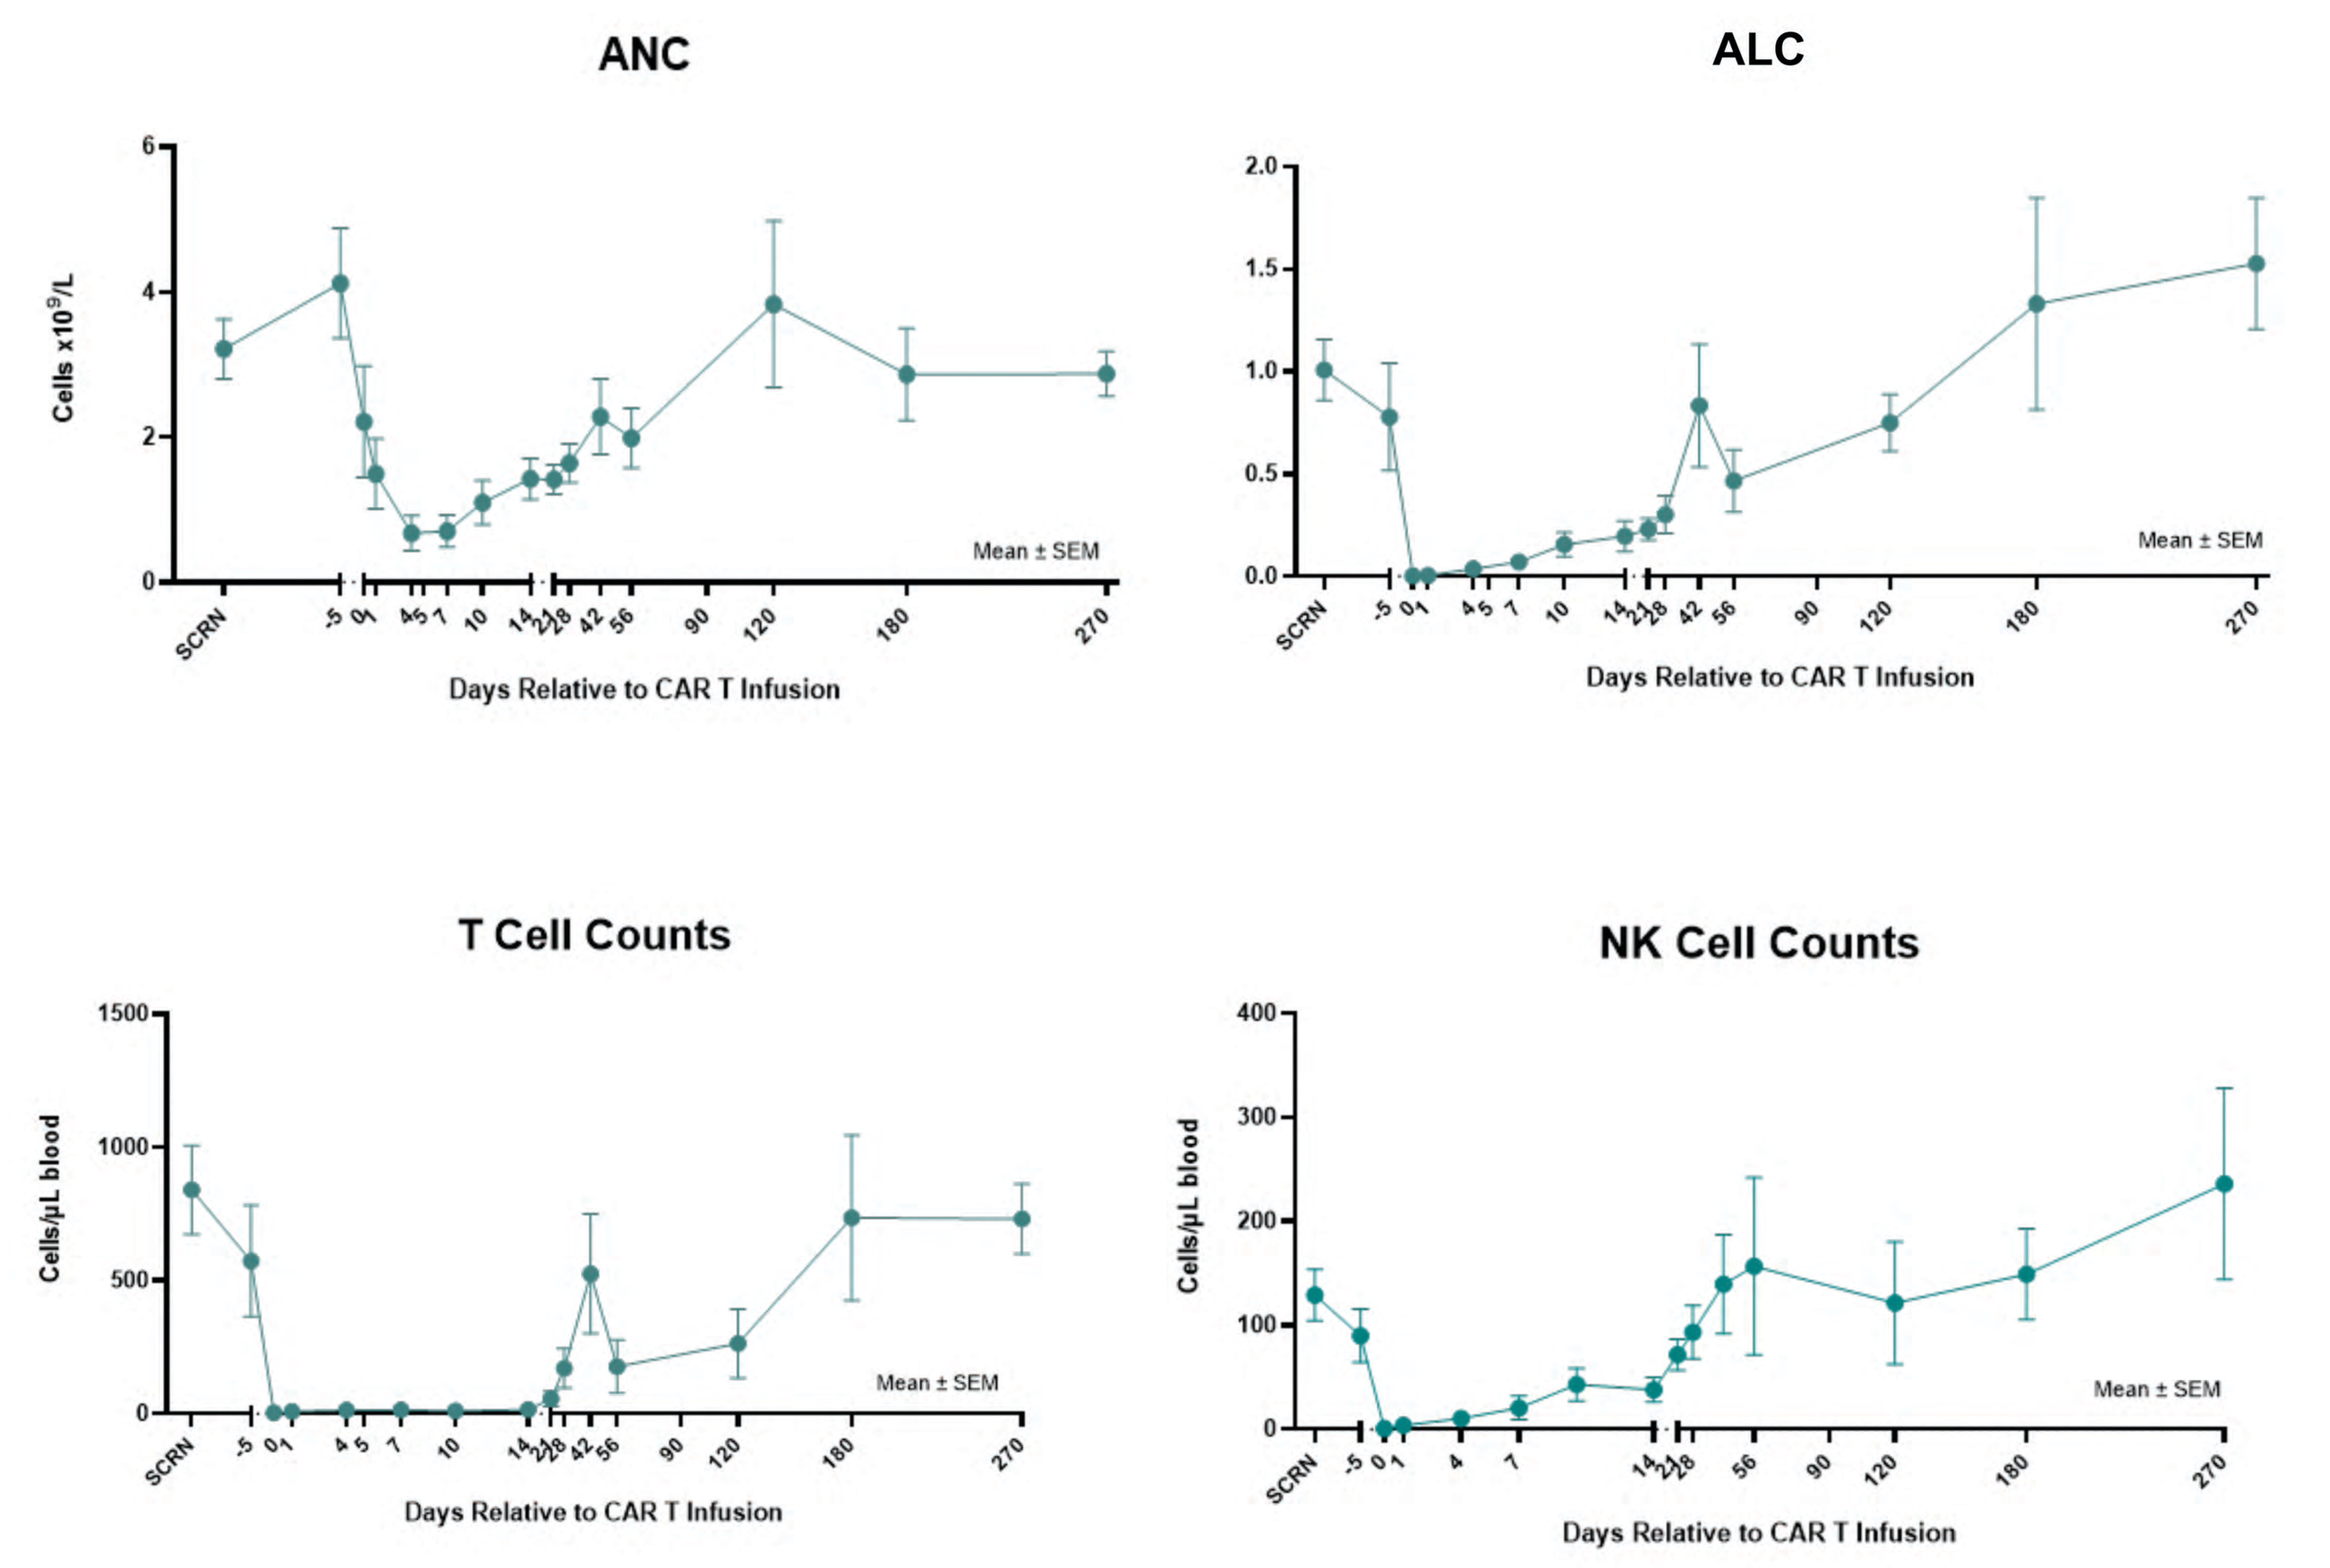

ANC
ALC
T Cell Counts
NK Cell Counts
Cells x10⁹/L
Cells/µL blood
Mean ± SEM
Days Relative to CAR T Infusion

ALC = absolute lymphocyte count; ANC = absolute neutrophil count; NK = natural killer.

Conclusions

- A single dose of ALLO-501/ALLO-501A following FCA90 provided durable remissions up to 31+ months that compared favorably to outcomes achieved with autologous CAR T cell therapies in patients with r/r LBCL.
- ALLO-501/ALLO-501A following FCA90 was generally well tolerated with only low-grade CRS, no ICANS, and no GvHD.
- Cytopenias and infections were manageable and comparable to experience with autologous CAR T cell therapies in r/r LBCL.
- Selective lymphodepletion with FCA90 creates a window for ALLO-501/501A engraftment, persistence, and anti-tumor activity.
- ALLO-501A, as an off-the-shelf, allogeneic CAR T cell product, eliminates the need for leukapheresis or bridging therapy, and may be more accessible to all eligible patients seeking CAR T therapy.
- These findings support broader evaluation of ALLO-501A with the selected Phase 2 regimen in the ongoing, first potentially pivotal Phase 2 trials (ALPHA2, NCT03939026, and EXPAND, NCT05714345) of an allogeneic CAR T cell product.

Acknowledgements: ALLO-501/ALLO-501A are anti-CD19 allogeneic CAR T (AlloCAR T™) therapies being jointly developed under a collaboration agreement between Servier and Allogene based on an exclusive license granted by Cellectis to Servier. ALLO-501/ALLO-501A uses Cellectis technologies. Servier grants to Allogene exclusive rights to ALLO-501/ALLO-501A in the U.S., while Servier retains exclusive rights for all other countries.

For questions or comments, please e-mail frederick.locke@moffitt.org.

Data Cutoff Date: April 20, 2023