ALLO-647 for Lymphodepletion in the Allogeneic CAR T Cell Setting: Safety Experience With ALLO-501/501A in Patients With Relapsed/Refractory Large B-Cell and Follicular Lymphomas

Poster #2095

Frederick L. Locke[1]; Javier L. Munoz[2]; Michael T. Tees[3]; Lazaros J. Lekakis[4]; Herbert A. Eradat[5]; Sven de Vos[5]; Rajneesh Nath[6]; Don A. Stevens[7]; Shahbaz A. Malik[8]; Geoffrey P. Shouse[9]; Mehdi Hamadani[10]; Olalekan O. Oluwole[11]; Miguel-Angel Perales[12]; David B. Miklos[13]; Anh Nguyen[14]; Amy Feng[14]; Lynn Navale[14]; Elaine Murray[14]; Greg P. Kaufman[14]; Kazuharu Kai[14]; John B. Le Gall[14]; Sattva S. Neelapu[15]

[1]Moffitt Cancer Center and Research Institute, Tampa, FL; [2]Mayo Clinic, Phoenix, AZ; [3]Colorado Blood Cancer Institute, Denver, CO; [4]University of Miami Health System, Sylvester Comprehensive Cancer Center, Miami, FL; [5]University of California, Los Angeles, Los Angeles, CA; [6]Banner MD Anderson Cancer Center, Scottsdale, AZ; [7]Norton Cancer Institute, Louisville, KY; [8]St. David's South Austin Medical Center, Austin, TX; [9]City of Hope National Medical Center, Duarte, CA; [10]Medical College of Wisconsin, Milwaukee, WI; [11]Vanderbilt University Medical Center, Nashville, TN; [12]Memorial Sloan Kettering Cancer Center, New York, NY; [13]Stanford University School of Medicine, Stanford, CA; [14]Allogene Therapeutics, San Francisco, CA; [15]The University of Texas MD Anderson Cancer Center, Houston, TX

BACKGROUND

- Allogeneic chimeric antigen receptor (CAR) T cell therapies derived from healthy donors may address limitations associated with autologous CAR T cell therapies by providing immediately available, off-the-shelf therapeutic options for patients with relapsed/refractory (R/R) large B-cell lymphoma (LBCL)[1,2]
- For allogeneic CAR T cells to be successful, there must be a safe and effective way to control host lymphocyte rejection of allogeneic CAR T cells (allo-rejection)[2,3]
- Lymphodepletion regimens including ALLO-647, a proprietary anti-CD52 monoclonal antibody, are designed to selectively prevent host rejection of *CD52*-knockout allogeneic CAR T cell products such as ALLO-501 and ALLO-501A (**Figure 1**)[4,5]

Figure 1. ALLO-501/501A and ALLO-647

Lymphodepletion Is Enhanced by Addition of ALLO-647, an Anti-CD52 Monoclonal Antibody

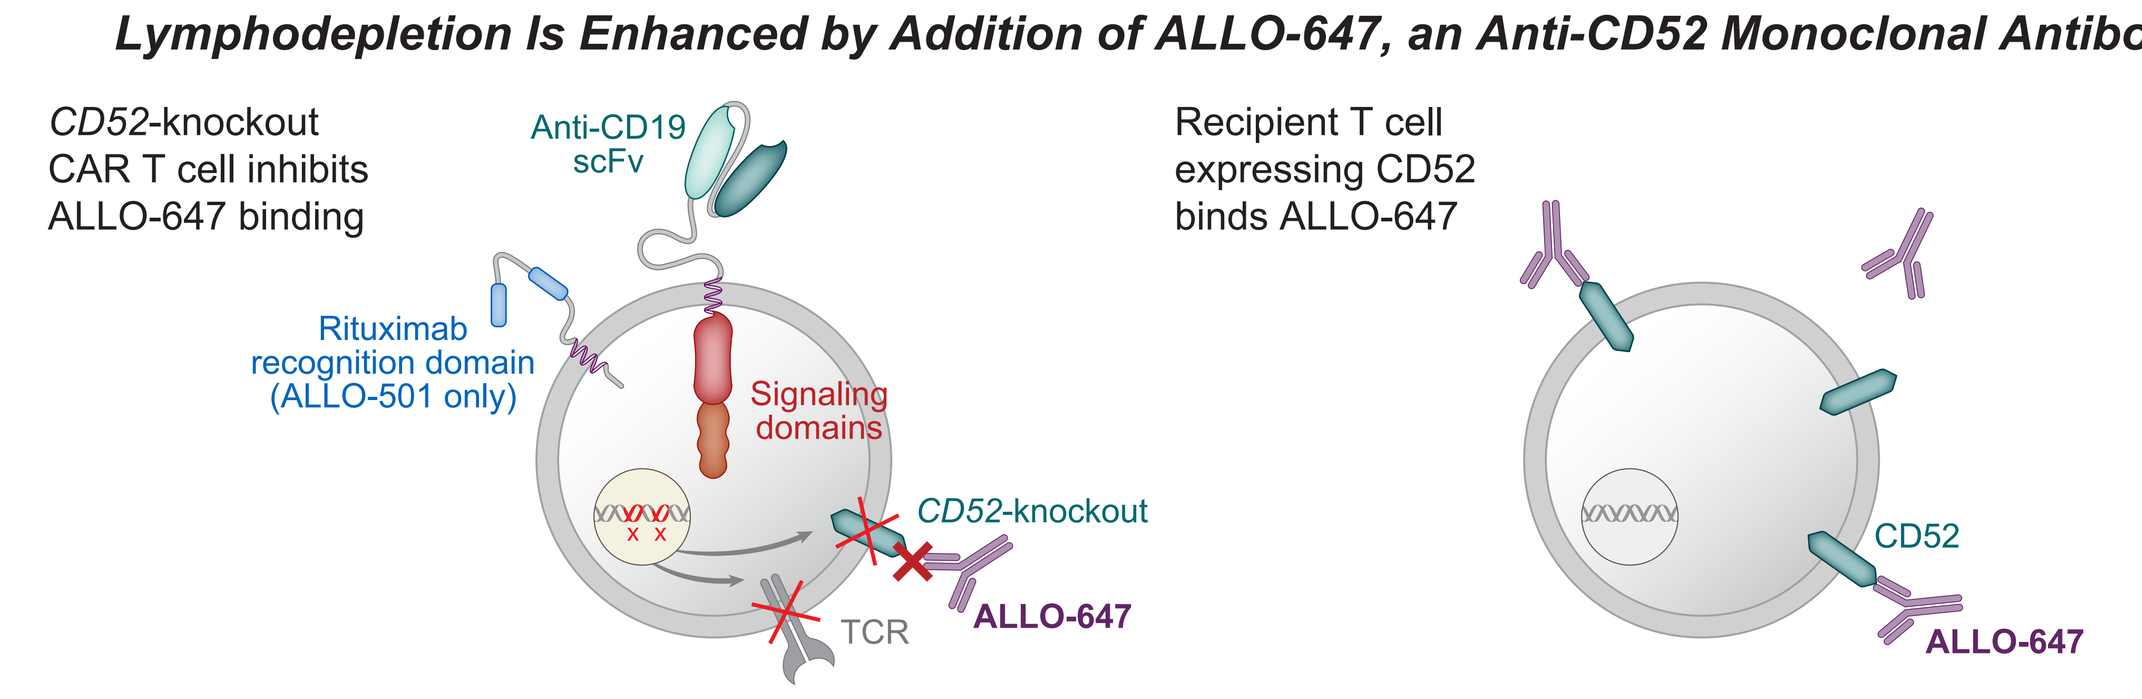

CD52-knockout CAR T cell inhibits ALLO-647 binding
Anti-CD19 scFv
Rituximab recognition domain (ALLO-501 only)
Signaling domains
CD52-knockout
TCR
ALLO-647
Recipient T cell expressing CD52 binds ALLO-647
CD52
ALLO-647

ALLO-501 and ALLO-501A CAR T cell products utilize TALEN® gene editing technology.
CAR, chimeric antigen receptor; scFV, single chain variable fragment; TCR, T cell receptor.

- Autologous CAR T cell therapy following lymphodepletion is associated with adverse events such as cytopenias and infections[6-9]
 - Prolonged grade ≥3 cytopenias are common after administration, occurring in up to 44% of patients at day 30
 - Grade ≥3 infections occurred in 12%-33% of patients
- In the phase 1 experience of patients with R/R LBCL (ALPHA and ALPHA2), lymphodepletion with standard fludarabine and cyclophosphamide (FC) and ALLO-647 followed by administration of allogeneic CD19 CAR T cell therapy provided durable responses and a manageable safety profile in 33 CAR T cell–naive patients[10,11]
 - After a median follow-up time of 27.7 months, overall response rate and complete remission rate was 58% and 42%, respectively, with a median duration of response of 23.1 months[11]
 - No graft-versus-host disease (GvHD), grade ≥3 cytokine release syndrome (CRS), or immune effector cell–associated neurotoxicity syndrome (ICANS) were observed; infections were primarily low grade and manageable[10,11]
- Initial evaluations showed that ALLO-647–containing lymphodepletion regimens had a tolerable safety profile[4]
- The objective of this analysis was to evaluate safety and pharmacokinetics/pharmacodynamics (PK/PD) effects of ALLO-647 in all 87 patients with R/R LBCL and follicular lymphoma (FL) from the ALPHA and ALPHA2 phase 1 studies

METHODS

STUDY DESIGN AND PATIENT POPULATION

- All treated patients from the ALPHA and ALPHA2 studies were included in this analysis
 - ALPHA (NCT0393026) is a phase 1 study evaluating ALLO-501 in adult patients with R/R LBCL or FL
 - ALPHA2 (NCT04416984) is a phase 1/2 study evaluating ALLO-501A in adult patients with R/R LBCL and FL grade 3b
- Patients treated in the studies received a lymphodepletion regimen of FC and ALLO-647 followed by administration of ALLO-501 or ALLO-501A (**Figure 2**)

Figure 2. Study Design

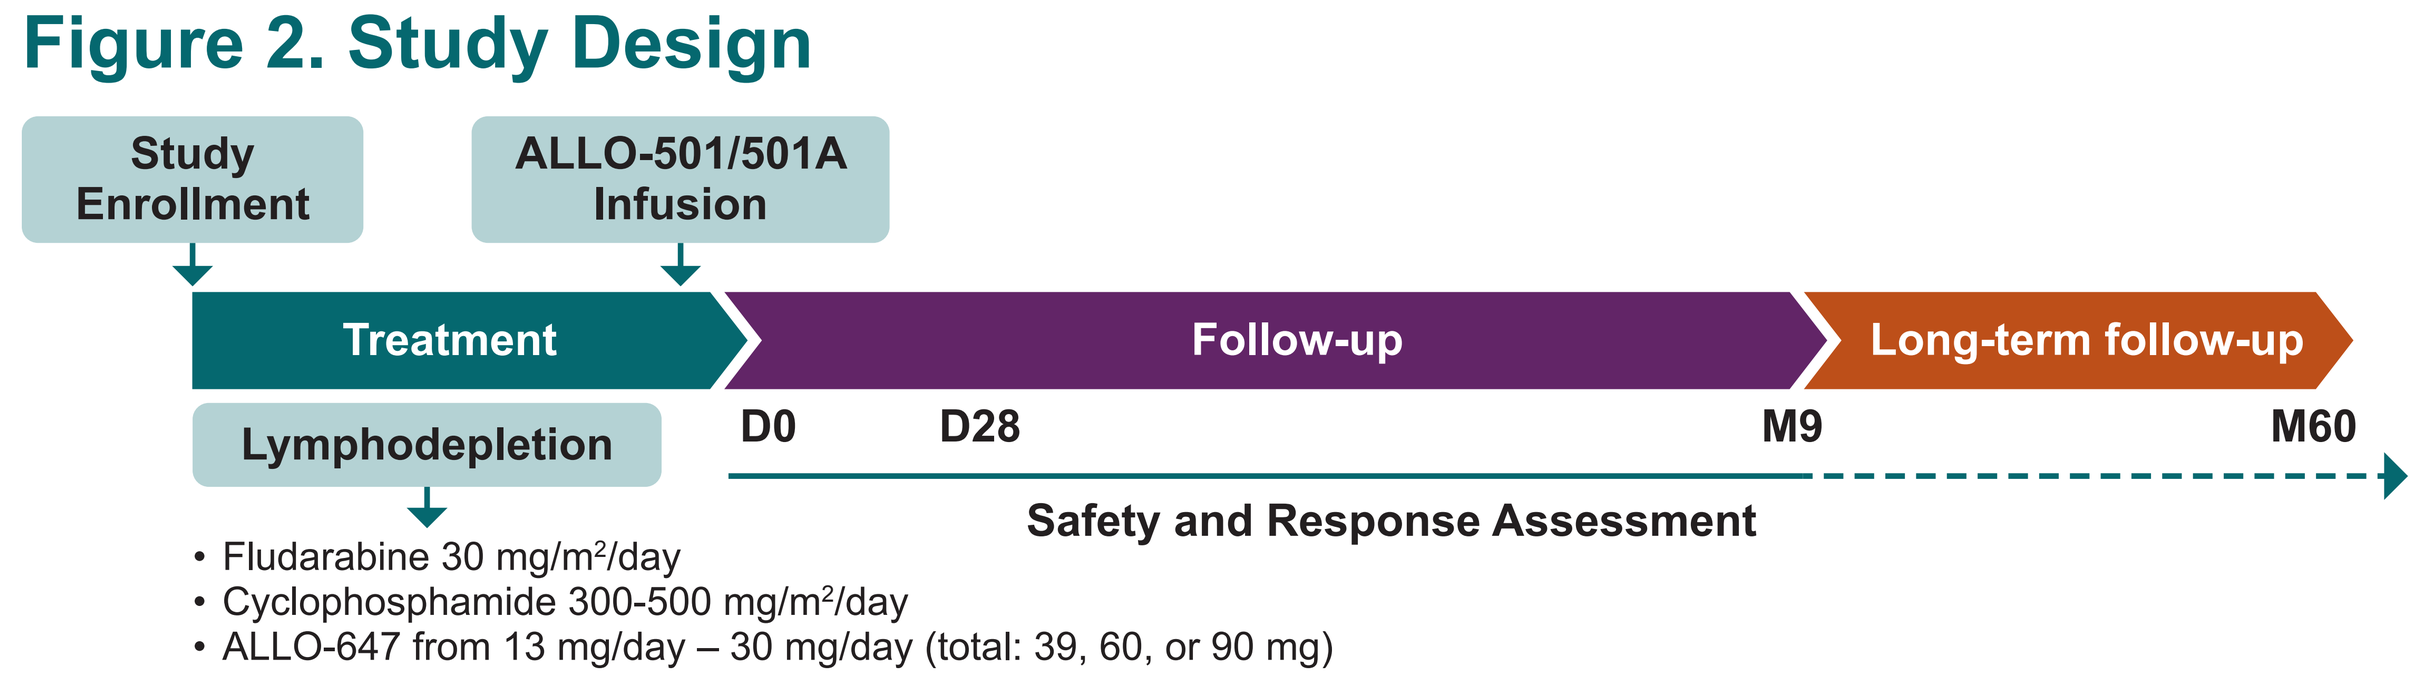

Study Enrollment
ALLO-501/501A Infusion
Treatment
Follow-up
Long-term follow-up
Lymphodepletion
D0
D28
M9
M60
Safety and Response Assessment
• Fludarabine 30 mg/m²/day
• Cyclophosphamide 300-500 mg/m²/day
• ALLO-647 from 13 mg/day – 30 mg/day (total: 39, 60, or 90 mg)

Regimens evaluated in phase 1 include: (1) LD followed by a single dose of ALLO-501/501A or (2) LD followed by a single dose of ALLO-501/501A and then an additional dose of ALLO-647 (30 mg) on D28 and ALLO-501/501A on D29.
D, day; LD, lymphodepletion; M, month.

ASSESSMENTS

- Safety of ALLO-647–containing lymphodepletion was assessed in all enrolled patients who received any study drug (safety analysis population)
 - Treatment-emergent adverse events (TEAEs) include any adverse events (AEs) occurring from the first dose of any study drug through the start of retreatment in the study, death, or the date prior to initiation of another anticancer agent, whichever comes first
 - Incidence rates of grade ≥3 cytopenias (neutropenia, thrombocytopenia, anemia, and pancytopenia) were assessed at study day 28, day 56, and month 4
 - Patients underwent weekly cytomegalovirus (CMV) monitoring via polymerase chain reaction until a minimum of 2 months after treatment
- PK/PD effects were examined to assess extent and effectiveness of lymphodepletion with ALLO-647 in all treated patients with an available assessment (PK/PD analysis population)
 - CAR T cell expansion was assessed in responders and nonresponders to ALLO-501/501A infusion
 - Leukocyte reconstitution was evaluated following ALLO-647–containing lymphodepletion and ALLO-501/501A infusion

RESULTS

PATIENTS

Figure 3. Patient Disposition

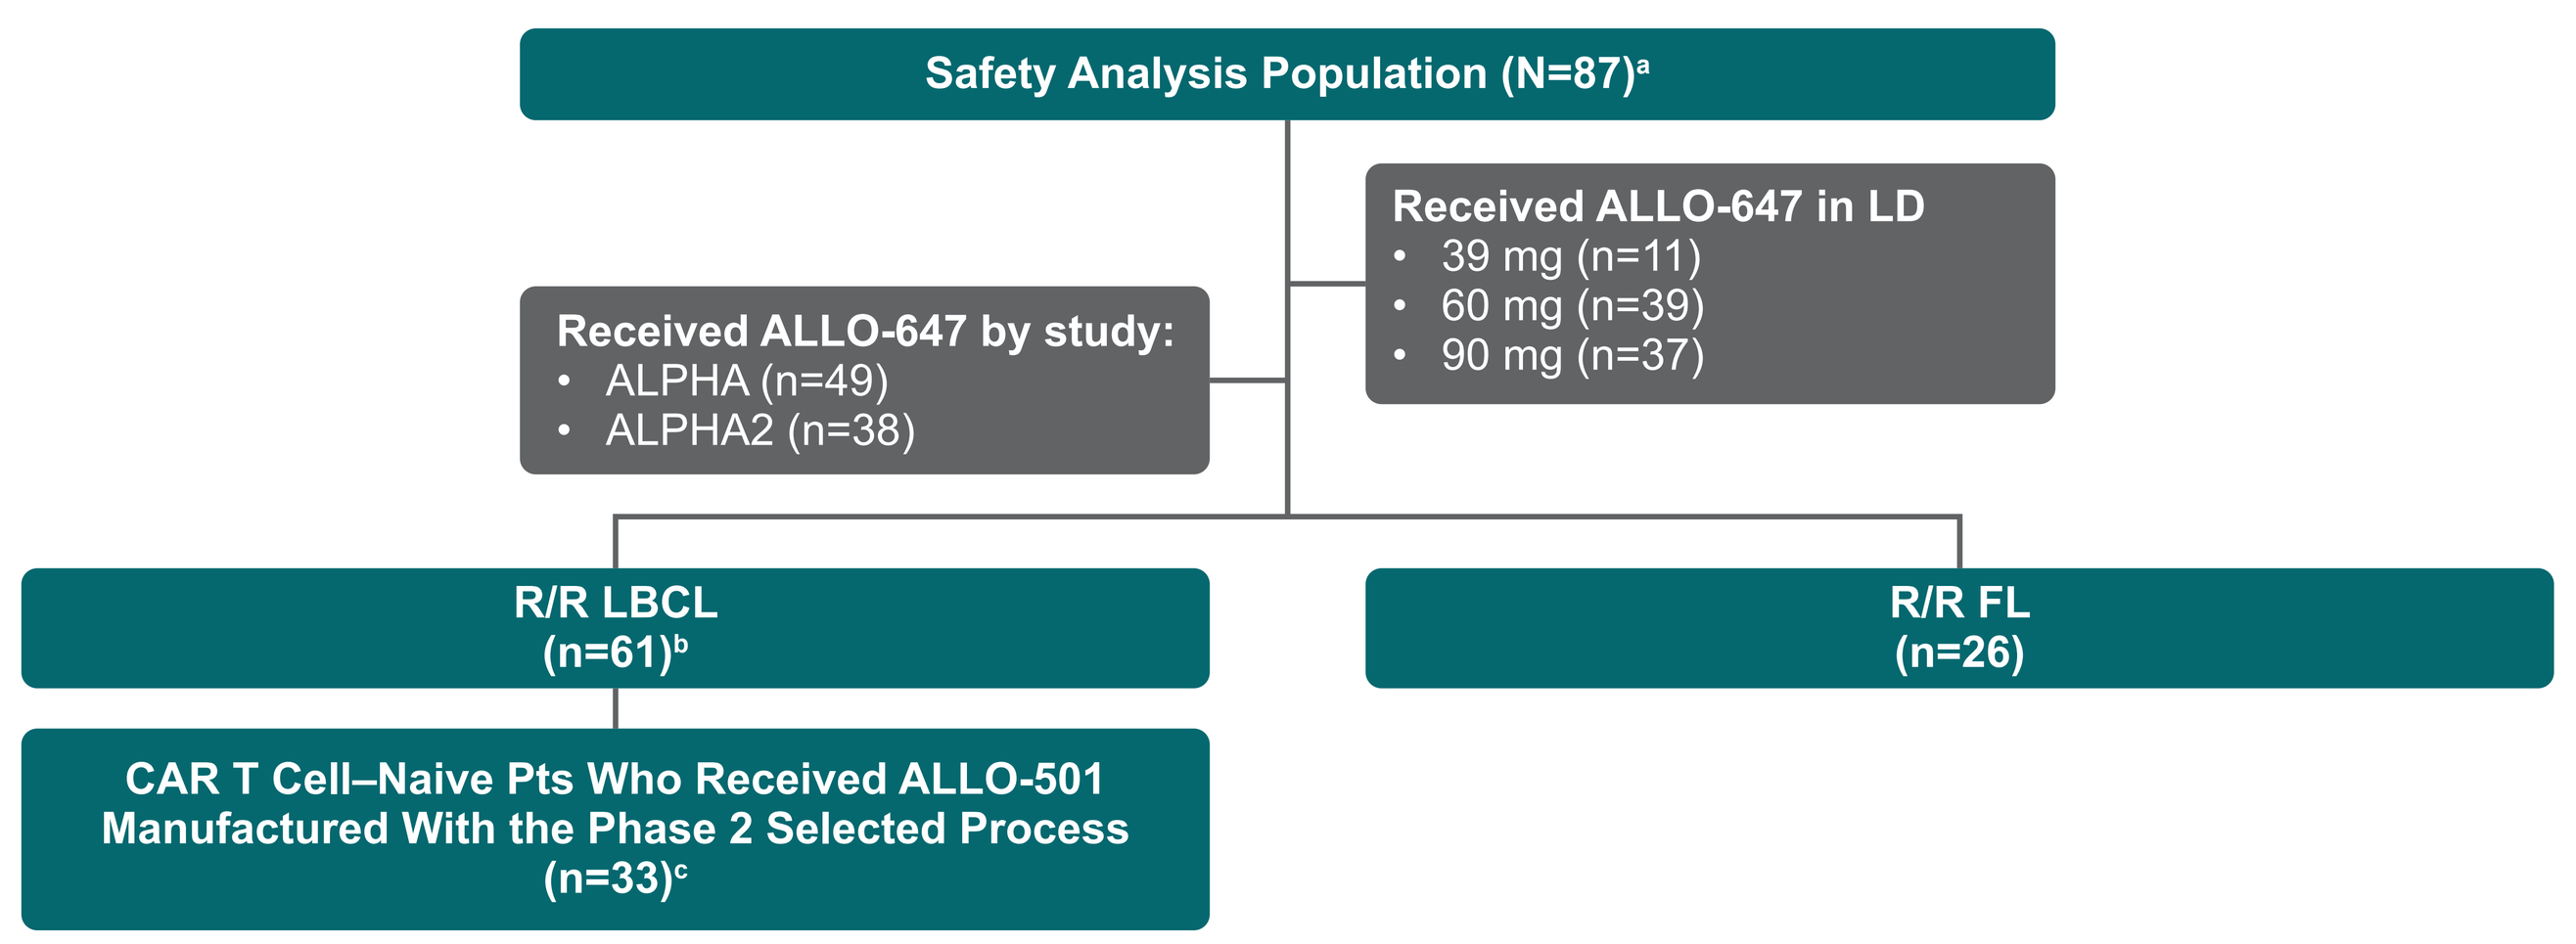

Safety Analysis Population (N=87)a
Received ALLO-647 in LD
• 39 mg (n=11)
• 60 mg (n=39)
• 90 mg (n=37)
Received ALLO-647 by study:
• ALPHA (n=49)
• ALPHA2 (n=38)
R/R LBCL (n=61)b
R/R FL (n=26)
CAR T Cell–Naive Pts Who Received ALLO-501 Manufactured With the Phase 2 Selected Process (n=33)c

Data cutoff date: April 20, 2023.
[a] All enrolled patients who received any study drug. [b] Includes 2 participants with grade 3b FL per study protocol. [c] Includes 1 participant with grade 3b FL per study protocol.
CAR, chimeric antigen receptor; FL, follicular lymphoma; LBCL, large B-cell lymphoma; LD, lymphodepletion; pt, patient; R/R, relapsed/refractory.

Table 1. Baseline Patient and Disease Characteristics

Characteristic	All (N=87)	LBCL: All LBCL (n=61)	LBCL: CAR T Cell–Naive Pts Who Received ALLO-501/501A Manufactured With the Phase 2 Selected Process (n=33)	FL (n=26)
Median age, years (range)	64 (31-77)	64 (31-76)	66 (31-76)	64 (34-77)
Stage IV disease, n (%)	51 (59)	40 (66)	19 (58)	11 (42)
ECOG PS 1, n (%)	61 (70)	48 (79)	26 (79)	13 (50)
Baseline LDH >ULN, n (%)	54 (62)	44 (72)	22 (67)	10 (38)
IPI score 3-5, n (%)	43 (49)	35 (57)	19 (58)	8 (31)
Germinal center subtype, n (%)	42 (48)	38 (62)	18 (55)	4 (15)
Double or triple hit, n (%)	23 (26)	20 (33)	10 (30)	3 (12)
Median number of prior regimens, n (range)	3 (2-12)	3 (2-9)	3 (2-8)	4 (2-12)
Prior autologous transplant, n (%)	6 (7)	6 (10)	3 (9)	0
Extranodal disease, n (%)	48 (55)	36 (59)	19 (58)	12 (46)

Data cutoff date: April 20, 2023.
CAR, chimeric antigen receptor; ECOG PS, Eastern Cooperative Oncology Group performance status; FL, follicular lymphoma; IPI, International Prognostic Index; LBCL, large B-cell lymphoma; LDH, lactate dehydrogenase; pt, patient; ULN, upper limit of normal.

SAFETY

- The most common any grade TEAEs included neutropenia (79%), anemia (61%), infusion-related reactions (IRRs, 55%), and thrombocytopenia (53%; **Table 2**)
- The most common grade ≥3 TEAEs included neutropenia (74%), anemia (38%), and thrombocytopenia (38%; **Table 2**)
- Grade ≥3 cytopenias decreased over time from day 28 (29%) to day 56 (20%) to month 4 (15%), which was consistent across subsets of patients with R/R LBCL and FL in this analysis and those previously observed[8] (**Table 3**)

Table 2. Safety – Most Common TEAEs

TEAEs, n (%)	All (N=87)	LBCL: All LBCL (n=61)	LBCL: CAR T Cell–Naive Pts Who Received ALLO-501/501A Manufactured With the Phase 2 Selected Process (n=33)	FL (n=26)
Any grade TEAEs				
Neutropenia	69 (79)	49 (80)	27 (82)	20 (77)
Anemia	53 (61)	42 (69)	23 (70)	11 (42)
Infusion-related reaction	48 (55)	31 (51)	16 (48)	17 (65)
Thrombocytopenia	46 (53)	35 (57)	19 (58)	11 (42)
Nausea	45 (52)	30 (49)	13 (39)	15 (58)
Grade ≥3 TEAEs				
Neutropenia	64 (74)	48 (79)	26 (79)	16 (62)
Anemia	33 (38)	29 (48)	15 (45)	4 (15)
Thrombocytopenia	33 (38)	27 (44)	13 (39)	6 (23)
Lymphopenia	27 (31)	19 (31)	10 (30)	8 (31)
White blood cell count decreased	22 (25)	19 (31)	10 (30)	3 (12)
Cytomegalovirus infection reactivation	8 (9)	7 (11)	4 (12)	1 (4)
Hypoxia	8 (9)	7 (11)	4 (12)	1 (4)

Data cutoff date: April 20, 2023.
Assessed in all enrolled patients who receive any study drug (safety analysis population). TEAEs include any adverse event occurring from the first dose of any study drug through the start of another treatment period, death, or the date prior to initiation of another anticancer agent, whichever comes first.
CAR, chimeric antigen receptor; FL, follicular lymphoma; LBCL, large B-cell lymphoma; pt, patient; TEAE, treatment-emergent adverse event.

Table 3. Safety – Grade ≥3 Cytopenias Over Time

Time	All (N=87)	LBCL: All LBCL (n=61)	LBCL: CAR T Cell–Naive Pts Who Received ALLO-501/501A Manufactured With the Phase 2 Selected Process (n=33)	FL (n=26)
Day 28, n (%)	25 (29)	20 (33)	11 (33)	5 (19)
Day 56, n (%)	17 (20)	14 (23)	7 (21)	3 (12)
Day 121 (Month 4), n (%)	13 (15)	11 (18)	6 (18)	2 (8)

Data cutoff date: April 20, 2023.
Assessed in all enrolled patients who receive any study drug (safety analysis population). TEAEs include any adverse event occurring from the first dose of any study drug through the start of another treatment period, death, or the date prior to initiation of another anticancer agent, whichever comes first. Grade ≥3 cytopenias include neutropenia, thrombocytopenia, anemia, or pancytopenia.
CAR, chimeric antigen receptor; FL, follicular lymphoma; LBCL, large B-cell lymphoma; pt, patient; TEAE, treatment-emergent adverse event.

- After treatment with ALLO-501/501A, 21 patients (24%) experienced CRS events, which were low grade except for 1 (1%) grade 3 event (**Table 4**)
- No GvHD or grade ≥3 ICANS were reported (**Table 4**)
- Infection events were primarily low grade and manageable with supportive care measures (**Tables 4 and 5**)
 - The most common infection was CMV reactivation (any grade, 25%; grade ≥3, 9%; **Table 5**)
 - Two patients (2%) had fatal infections (COVID-19 pneumonia and pneumonia, n=1 each), both in the FL subgroup (**Table 5**)
- In total, 8 patients (9%) experienced fatal AEs (respiratory failure, n=1; B-cell lymphoma, n=2; torsade de pointes, n=1; cerebrovascular accident, n=1; multiple organ dysfunction syndrome, n=1; multiple organ dysfunction syndrome and pneumonia, n=1; COVID-19 pneumonia, n=1)
 - None of the grade 5 AEs were considered related to ALLO-647 or ALLO-501/501A
- Addition of ALLO-647 to the standard FC lymphodepletion regimen did not result in AEs beyond those commonly observed in autologous CAR T cell therapy[6,7]

Table 4. Safety – TEAEs of Special Interest

TEAEs, n (%)	All (N=87) Any Grade	All (N=87) Grade ≥3	LBCL: All LBCL (n=61) Any Grade	LBCL: All LBCL (n=61) Grade ≥3	LBCL: CAR T Cell–Naive Pts Who Received ALLO-501/501A Manufactured With the Phase 2 Selected Process (n=33) Any Grade	LBCL: CAR T Cell–Naive Pts (n=33) Grade ≥3	FL (n=26) Any Grade	FL (n=26) Grade ≥3
CRS	21 (24)	1 (1)	17 (28)	1 (2)	8 (24)	0	4 (15)	0
ICANS	1 (1)	0	1 (2)	0	0	0	0	0
Neurotoxicity	24 (28)	3 (3)	19 (31)	3 (5)	13 (39)	2 (6)	5 (19)	0
GvHD	0	0	0	0	0	0	0	0
IRR	48 (55)	5 (6)	31 (51)	4 (7)	16 (48)	3 (9)	17 (65)	1 (4)
Infections	50 (57)	18 (21)	34 (56)	13 (21)	19 (58)	5 (15)	16 (62)	5 (19)

Data cutoff date: April 20, 2023.
Assessed in all enrolled patients who receive any study drug (safety analysis population). TEAEs include any adverse event occurring from the first dose of any study drug through the start of another treatment period, death, or the date prior to initiation of another anticancer agent, whichever comes first.
Grade ≥3 neurotoxicity events were muscular weakness (2%) and encephalopathy (1%).
CAR, chimeric antigen receptor; CRS, cytokine release syndrome; FL, follicular lymphoma; GvHD, graft-versus-host disease; ICANS, immune effector cell-associated neurotoxicity syndrome; IRR, infusion-related reaction; LBCL, large B-cell lymphoma; pt, patient; TEAE, treatment-emergent adverse event.

Table 5. Safety – Most Common Infections (≥5% Any Grade)

TEAEs, n (%)	All (N=87) Any Grade	All (N=87) Grade ≥3	LBCL: All LBCL (n=61) Any Grade	LBCL: All LBCL (n=61) Grade ≥3	LBCL: CAR T Cell–Naive Pts Who Received ALLO-501/501A Manufactured With the Phase 2 Selected Process (n=33) Any Grade	LBCL: CAR T Cell–Naive Pts (n=33) Grade ≥3	FL (n=26) Any Grade	FL (n=26) Grade ≥3
Viral infections	34 (39)	12 (14)	22 (36)	9 (15)	13 (39)	4 (12)	12 (46)	3 (12)[a]
CMV	22 (25)	8 (9)	16 (26)	7 (11)	10 (30)	4 (12)	6 (23)	1 (4)
COVID-19	5 (6)	1 (1)	2 (3)	1 (2)	2 (6)	1 (3)	3 (12)	0
Other infections	25 (29)	12 (14)[b]	16 (26)	8 (13)	8 (24)	5 (15)	9 (35)	4 (15)[b]
Pneumonia	8 (9)	6 (7)[b]	4 (7)	3 (5)	4 (12)	3 (9)	4 (15)	3 (12)[b]
Sepsis	5 (6)	5 (6)	4 (7)	4 (7)	2 (6)	2 (6)	1 (4)	1 (4)
Bacterial infections	10 (11)	4 (5)	9 (15)	4 (7)	3 (9)	2 (6)	1 (4)	0
Fungal infections	7 (8)	2 (2)	5 (8)	1 (2)	2 (6)	0	2 (8)	1 (4)

Data cutoff date: April 20, 2023.
Assessed in all enrolled patients who receive any study drug (safety analysis population). TEAEs include any adverse event occurring from the first dose of any study drug through the start of another treatment period, death, or the date prior to initiation of another anticancer agent, whichever comes first.
[a] Includes a grade 5 event (COVID-19 pneumonia). [b] Includes a grade 5 event (pneumonia).
CAR, chimeric antigen receptor; CMV, cytomegalovirus; pt, patient; TEAE, treatment-emergent adverse event.

PHARMACOKINETICS

- ALLO-647 showed a dose-dependent increase in exposure following ALLO-501/501A administration (**Figure 4A**)
- CAR T cell expansion was observed following ALLO-647–containing lymphodepletion, which was superior in responders versus nonresponders with CAR T cell persistence up to month 4 (**Figure 4B**); responders generally received a higher dose of ALLO-647 than nonresponders (data not shown)
- Following lymphodepletion, the median time to absolute neutrophil count and absolute lymphocyte count recovery to grade <4 was 7 days and 28 days, respectively (**Figures 5A and 5B**)

Figure 4. ALLO-647 Exposure (A) and CAR T Cell Expansion (B)

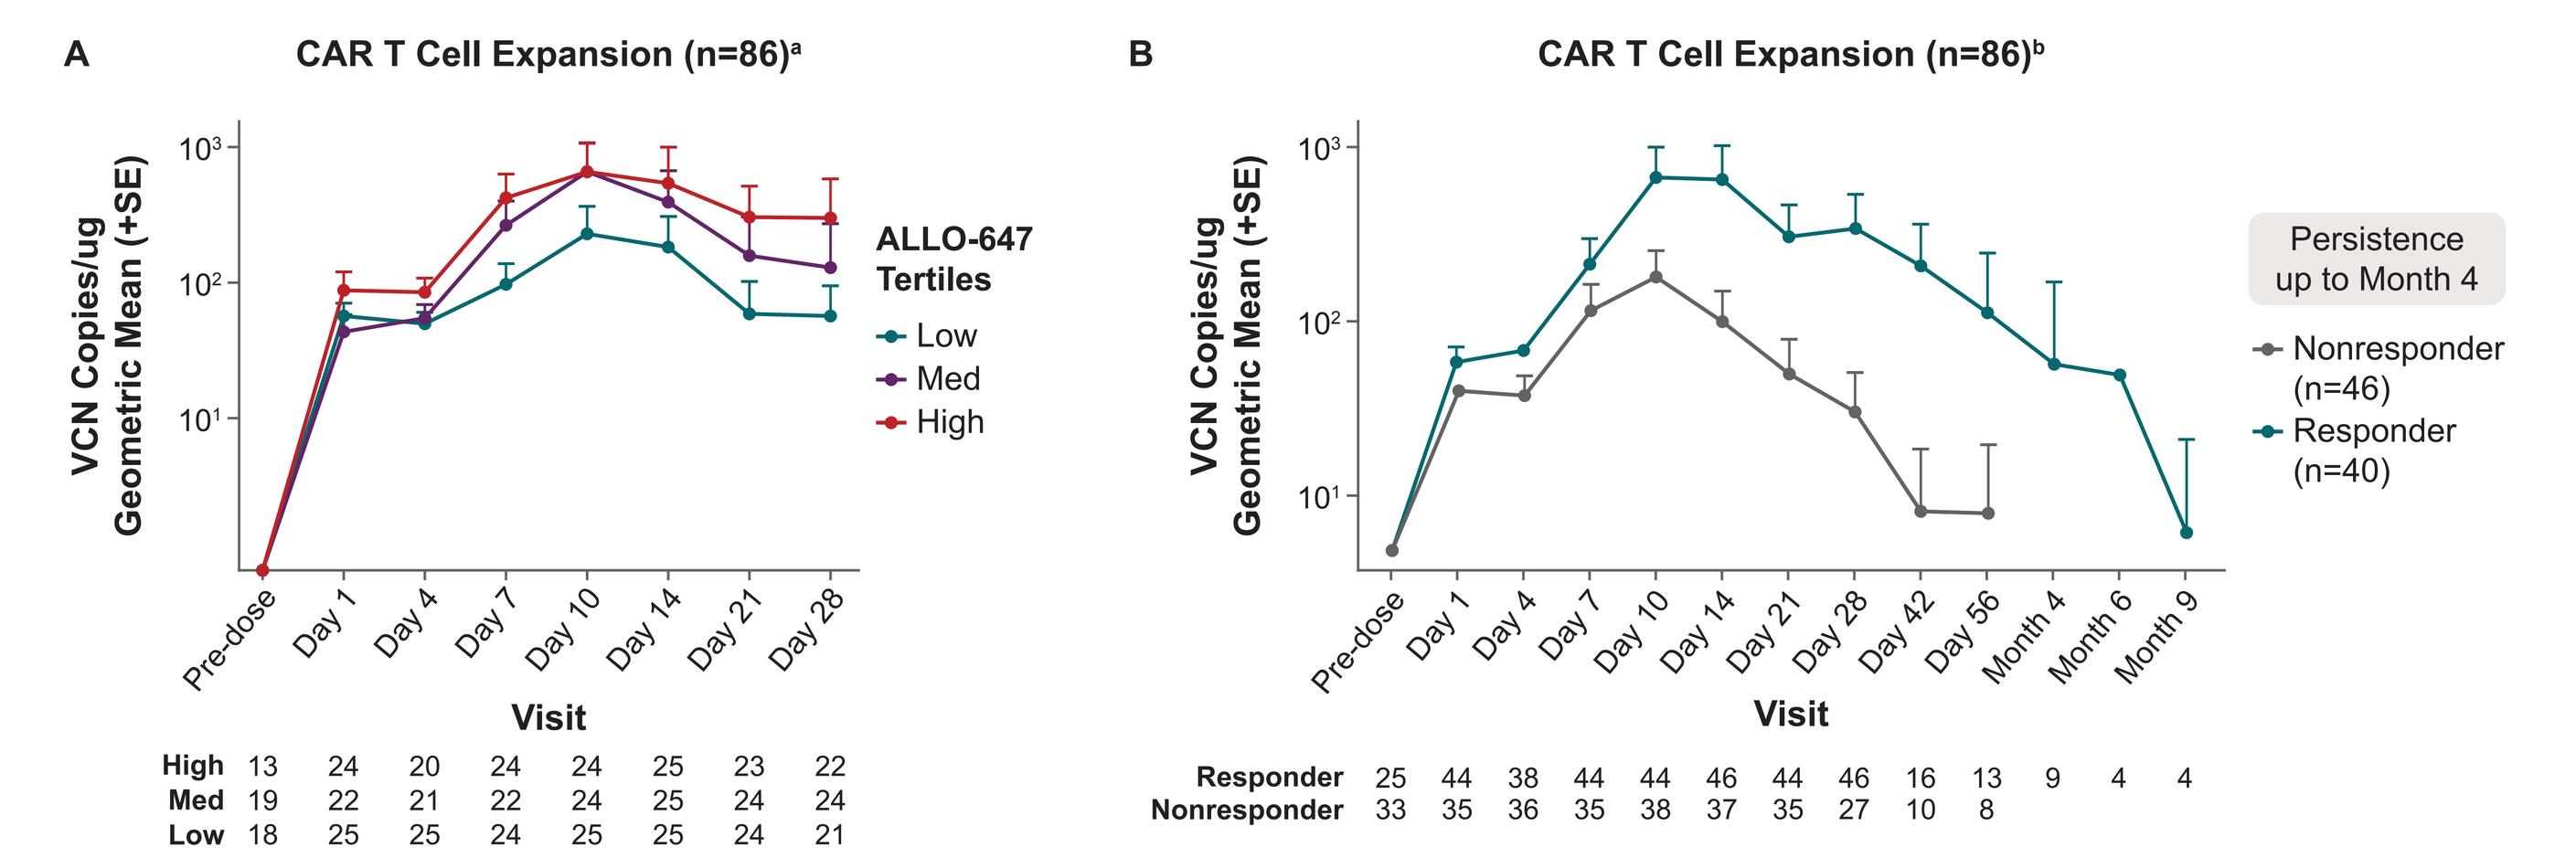

A
CAR T Cell Expansion (n=86)a
VCN Copies/ug Geometric Mean (+SE)
10¹
10²
10³
ALLO-647 Tertiles
Low
Med
High
Pre-dose Day 1 Day 4 Day 7 Day 10 Day 14 Day 21 Day 28
Visit
High 13 24 20 24 24 25 23 22
Med 19 22 21 22 24 25 24 24
Low 18 25 25 24 25 25 24 21
B
CAR T Cell Expansion (n=86)b
VCN Copies/ug Geometric Mean (+SE)
10¹
10²
10³
Persistence up to Month 4
Nonresponder (n=46)
Responder (n=40)
Pre-dose Day 1 Day 4 Day 7 Day 10 Day 14 Day 21 Day 28 Day 42 Day 56 Month 4 Month 6 Month 9
Visit
Responder 25 44 38 44 44 46 44 46 16 13 9 4 4
Nonresponder 33 35 36 35 38 37 35 27 10 8

Data cutoff: April 20, 2023.
Assessed in all treated patients with an available assessments.
[a] Includes patients with 3-day ALLO-647 lymphodepletion; alternate dosing strategies were excluded. [b] Includes patients treated with ALLO-501/501A.
CAR, chimeric antigen receptor; med, medium; SE, standard error; VCN, vector copy number.

Figure 5. Recovery of Leukocyte Counts Over Time: ANC (A), ALC (B), and Count T Cells (C)

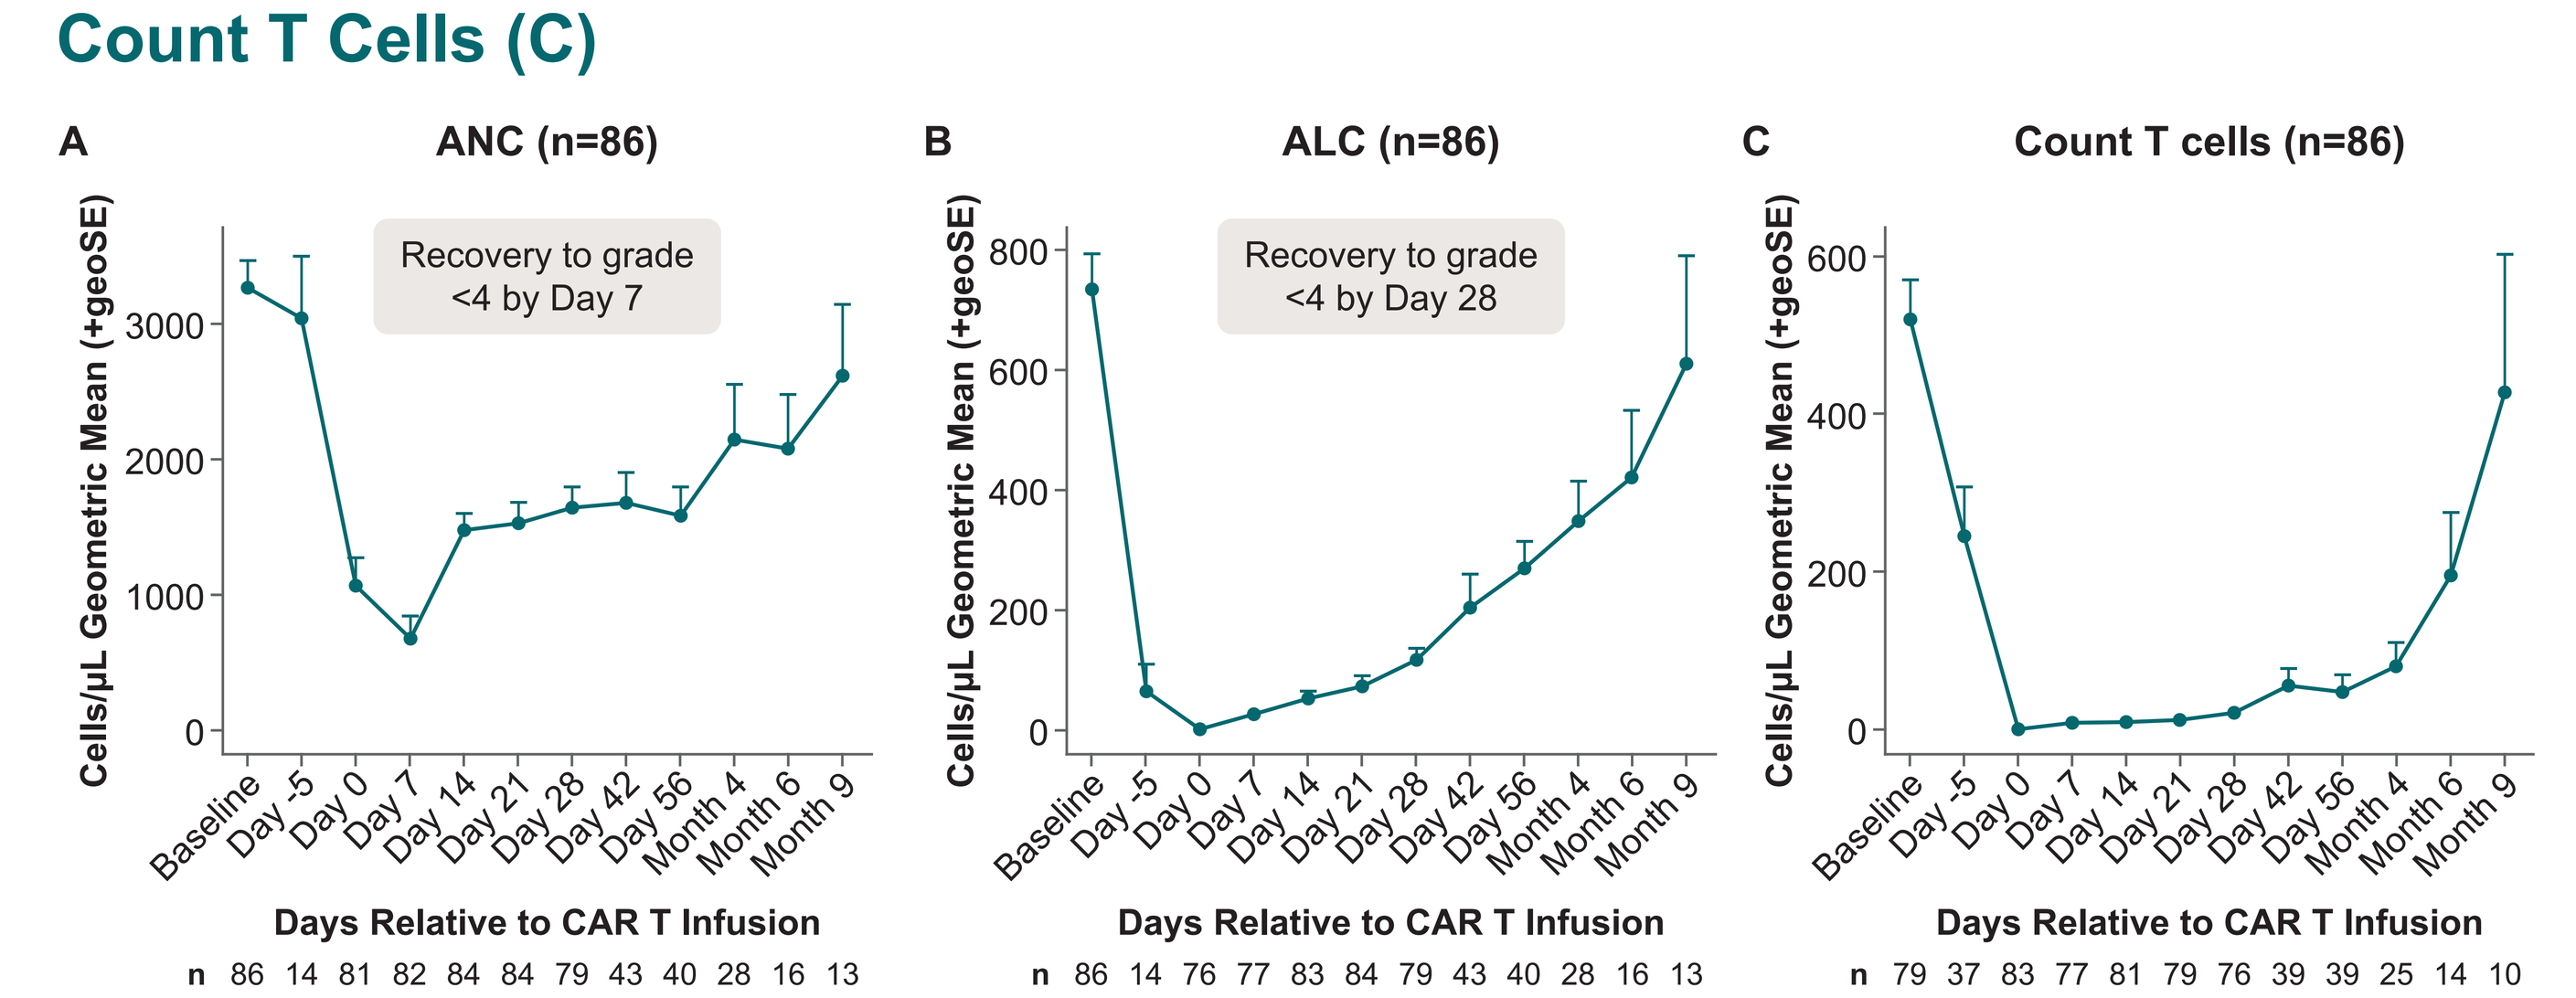

A
ANC (n=86)
Recovery to grade <4 by Day 7
Cells/µL Geometric Mean (+geoSE)
0
1000
2000
3000
B
ALC (n=86)
Recovery to grade <4 by Day 28
0
200
400
600
800
C
Count T cells (n=86)
0
200
400
600
Baseline Day -5 Day 0 Day 7 Day 14 Day 21 Day 28 Day 42 Day 56 Month 4 Month 6 Month 9
Days Relative to CAR T Infusion
n 86 14 81 82 84 84 79 43 40 28 16 13
n 86 14 76 77 83 84 79 43 40 28 16 13
n 79 37 83 77 81 79 76 39 39 25 14 10

Data cutoff: April 20, 2023.
Assessed in all treated patients with an available assessments.
ALC, absolute lymphocyte count; ANC, absolute neutrophil count; CAR, chimeric antigen receptor; geoSE, geometric standard error.

CONCLUSIONS

- In the phase 1 experience, the addition of ALLO-647 to standard lymphodepletion does not alter the safety profile. ALLO-647–enhanced lymphodepletion had a manageable safety profile consistent with that of standard lymphodepletion used with autologous CAR T cell therapy, including the incidence of cytopenias and infections[6-9]
- Optimized ALLO-647 exposure provides a window for expansion and persistence of *CD52*-knockout CD19 allogeneic CAR T cells
- The contribution of ALLO-647 to standard FC lymphodepletion regarding expansion, persistence, and improved clinical outcomes of ALLO-501A will be further evaluated in the ALPHA2 and EXPAND studies (NCT04416984 and NCT05714345)

REFERENCES

1. Lin H, et al. *Front Immunol*. 2021;12:744823.
2. Lv Z, et al. *Front Immunol*. 2023;14:1199145.
3. Martínez Bedoya D, et al. *Front Immunol*. 2021;12:640082.
4. Tees MT, et al. *J Clin Oncol*. 2021;39(suppl 15):2527-2527.
5. Lekakis LJ, et al. *Blood*. 2021;138(suppl 1):649-649.
6. Logue JM, et al. *Haematologica*. 2021;106(4):978-986.
7. YESCARTA. Prescribing information. Kite Pharma, Inc.; 2022.
8. KYMRIAH. Prescribing Information. Novartis Pharmaceuticals Corp.; 2022.
9. BREYANZI. Prescribing Information. Juno Therapeutics, Inc.; 2022.
10. Munoz J, et al. *HemaSphere*. 2023;7(S3):e8808264.
11. Locke FL, et al. ICML 2023. Oral #048.

ACKNOWLEDGMENTS

ALLO-501 and 501A are allogeneic CAR T (AlloCAR T™) products that use Cellectis technology. ALLO-501/501A are CD19 AlloCAR T products being jointly developed under a collaboration agreement between Servier and Allogene based on an exclusive license granted by Cellectis to Servier. Servier grants to Allogene exclusive rights to ALLO-501/501A in the U.S. while Servier retains exclusive rights for all other countries.

This presentation is the intellectual property of the author/presenter.
Contact them at Frederick.Locke@moffitt.org for permission to reprint and/or distribute.

Presented at the American Society of Hematology Annual Meeting – December 9-12, 2023