Preclinical Evaluation of ALLO-329: Allogeneic CD19 CAR T Cells Expressing an Anti-Rejection CD70 CAR for the Treatment of Autoimmune Diseases

Poster #1841

Kristen Zhang[1], Joanne Li[1], Duy Nguyen[1], Nguyen Tan[1], Adam Mealy[1], Hsin-Yuan Cheng[1], David Huang[1], Suhasni Gopalakrishnan[1], Zachary Roberts[1], Cesar Sommer[1], Elvin Lauron[1]

[1]*Allogene Therapeutics, San Francisco, CA, United States*

ABSTRACT

Background/Purpose: Autologous CD19 chimeric antigen receptor (CAR) T cell therapies have recently shown to be well tolerated and highly effective in patients with autoimmune diseases (AID) including systemic lupus erythematosus (SLE), idiopathic inflammatory myositis, and systemic sclerosis. However, autologous CAR T cell therapies have major limitations such as complex logistics and manufacturing constraints that may hinder widespread access in these large patient populations. These challenges may be overcome with off-the-shelf allogeneic CAR T cell products derived from healthy donor T cells. Although allogeneic CAR T cells can provide immediate availability to patients and scalable manufacturing, they are more susceptible to rejection by the host immune system and therefore may have reduced persistence, limiting clinical responses. To address this challenge, we developed an anti-rejection CD70 CAR capable of selectively depleting activated (CD70⁺) alloreactive lymphocytes. This CD70 CAR may render allogeneic CD19 CAR T cells resistant to allorejection while endowing dual targeting of CD19⁺ B cells and CD70⁺ T cells. A CD19/CD70 dual CAR T cell therapy may therefore reduce or eliminate the need for lymphodepleting conditioning regimens that are typically required prior to CAR T cell infusion. Furthermore, because CD70 itself has been shown to be upregulated on T cells in AID, this approach could provide additional benefit to patients through elimination of CD70⁺ autoreactive T cells.

Methods: CRISPR gene-editing technology (ABR-001) combined with adeno-associated virus (AAV) transduction was employed to knock-in a bicistronic, CD19/CD70 dual CAR construct into the T Cell Receptor Alpha Constant (*TRAC*) locus. Following expansion, dual CAR T cells were characterized by flow cytometry analysis of CAR expression as well as markers of T cell activation and differentiation. Cytotoxicity and expansion of the dual CAR T cells, referred to as ALLO-329, were assessed *in vitro* and *in vivo* using B cell lymphoma models and humanized mouse models. The susceptibility of ALLO-329 to rejection by HLA-mismatched T cells was assessed in mixed lymphocyte reaction (MLR) assays.

Results: ALLO-329 exhibited cytotoxic activity and target-mediated expansion in the presence of tumor cell lines expressing CD19 and/or CD70. Primary CD19⁺ B cells and CD70⁺ T cells from healthy donors and patients with SLE were rapidly eliminated by ALLO-329 *in vitro* and *in vivo*, both in mouse peripheral blood and spleen, resulting in a concomitant reduction of antibody production. In *in vitro* MLR assays, ALLO-329 eliminated CD70⁺ alloreactive T cells and demonstrated resistance to rejection and longer persistence relative to CD19 CAR T cells, which were rapidly depleted.

Conclusions: ALLO-329 can be successfully produced via ABR-001-mediated site-specific integration of a single bicistronic construct in the *TRAC* locus, successfully yielding high numbers of CAR⁺ cells that show specific cytotoxic activity, rejection avoidance, and the ability to eradicate B cells in mouse tissues. This novel off-the-shelf allogeneic CD70/CD19 CAR T cell product is a promising candidate for clinical evaluation.

Figure 1. ALLO-329: Allogeneic CD19/CD70 Dual CAR T Cells

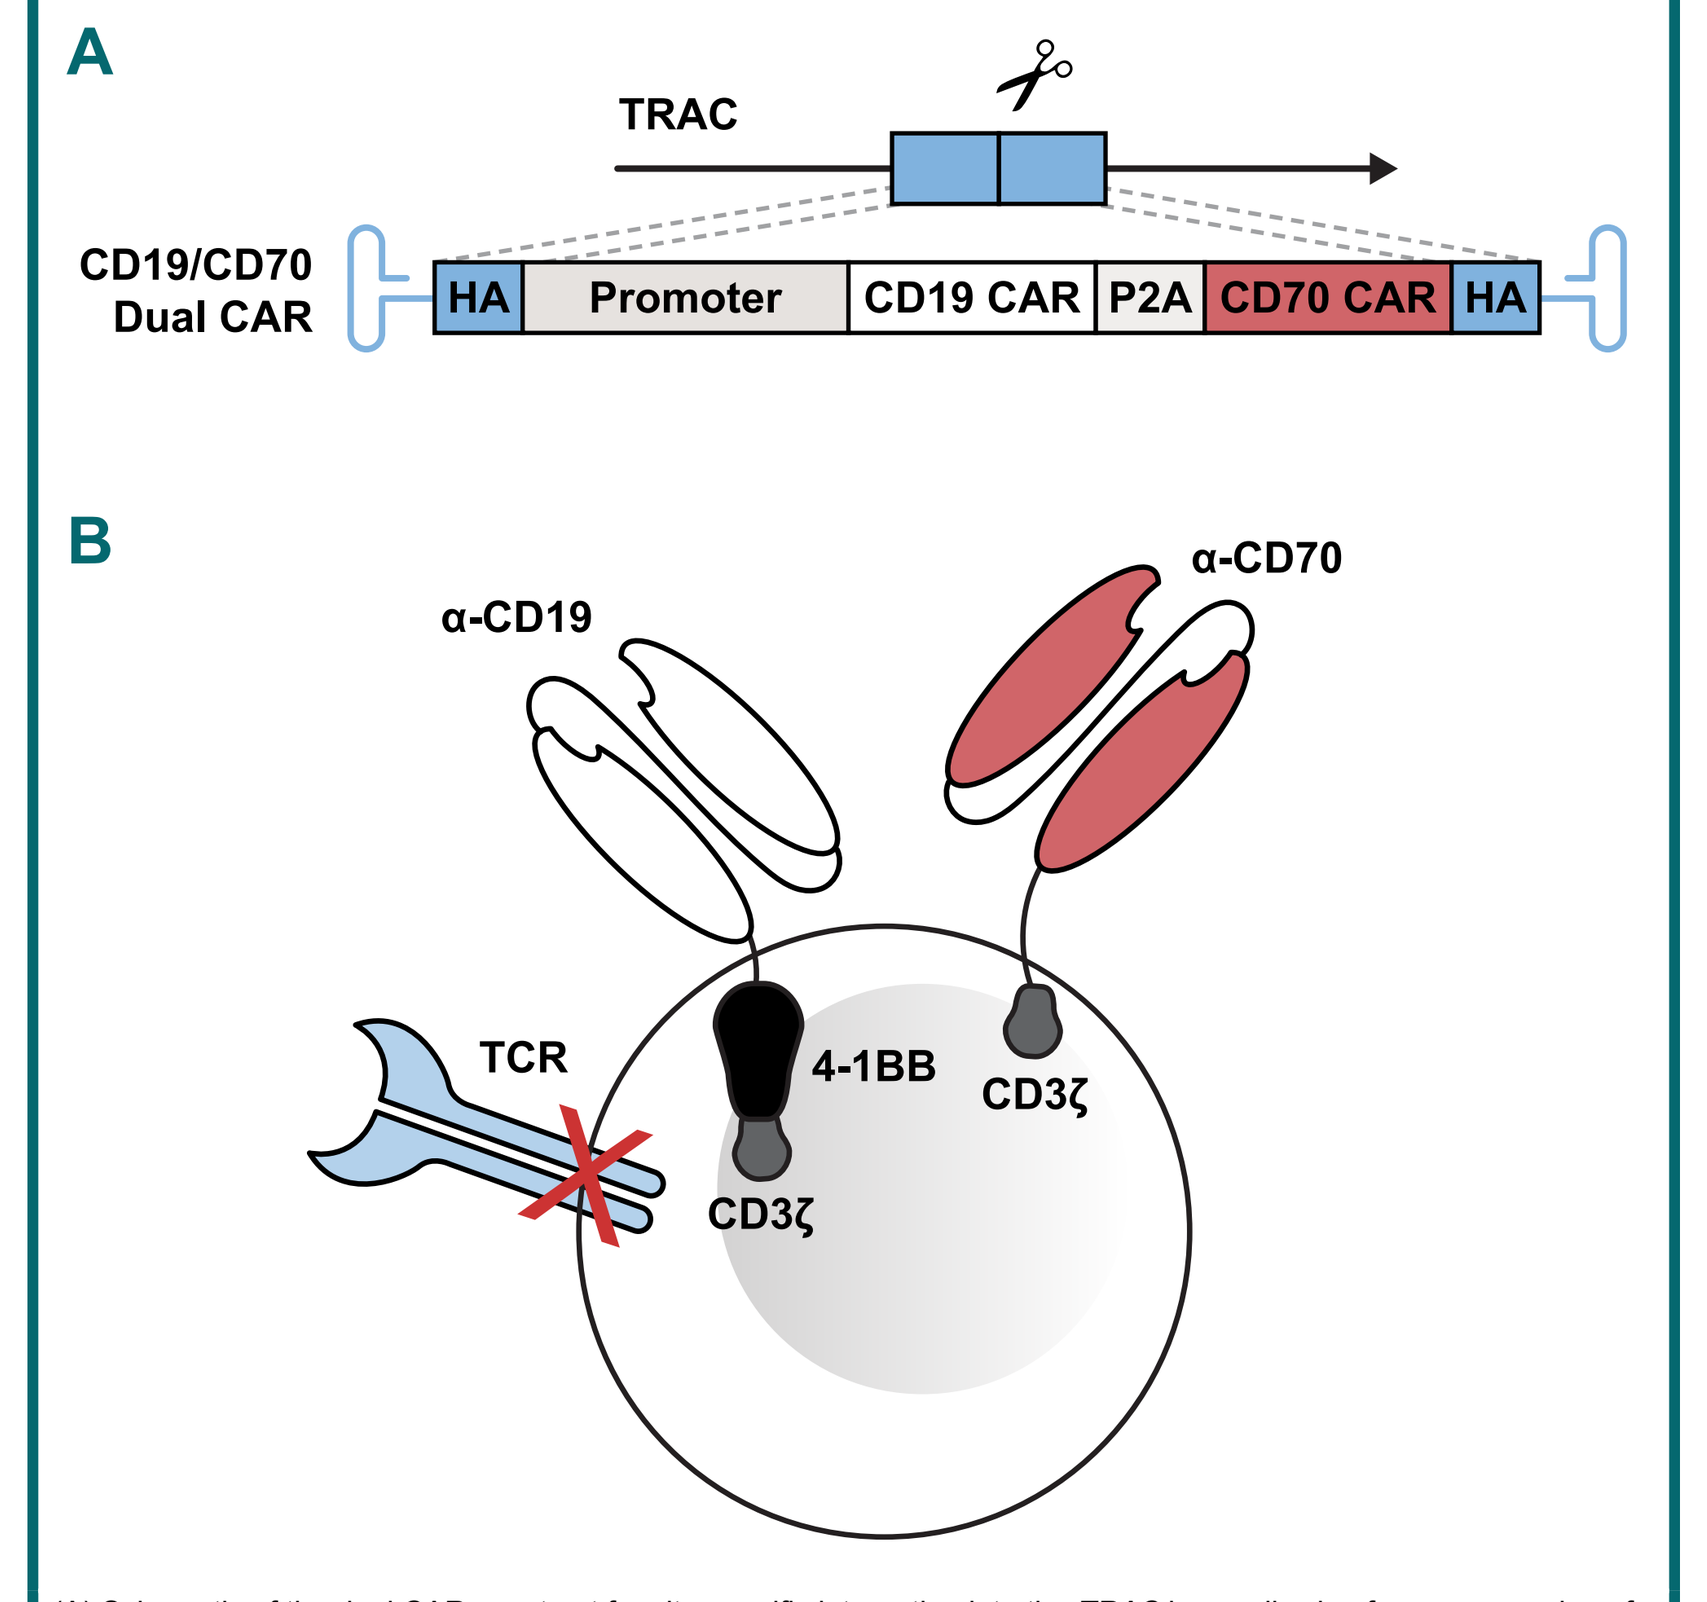
A
TRAC
CD19/CD70 Dual CAR
HA
Promoter
CD19 CAR
P2A
CD70 CAR
HA
B
α-CD19
α-CD70
TCR
4-1BB
CD3ζ
CD3ζ

(A) Schematic of the dual CAR construct for site-specific integration into the *TRAC* locus allowing for co-expression of a CD19 CAR and a CD70 anti-rejection CAR. (B) The CD19 CAR contains an scFv, a CD8α hinge and transmembrane domain and intracellular domains of 4-1BB (costimulatory domain) and CD3ζ (activation domain), while the CD70 CAR contains an scFv, a CD8α hinge and transmembrane domain, and the CD3ζ domain only without a costimulatory domain.
CAR, chimeric antigen receptor; scFv, single chain variable fragment; TCR, T cell receptor.

RESULTS

Figure 2. High Levels of CD70 CAR and CD19 CAR Expression Can Be Achieved via Site-Specific Integration of the Dual CAR Construct

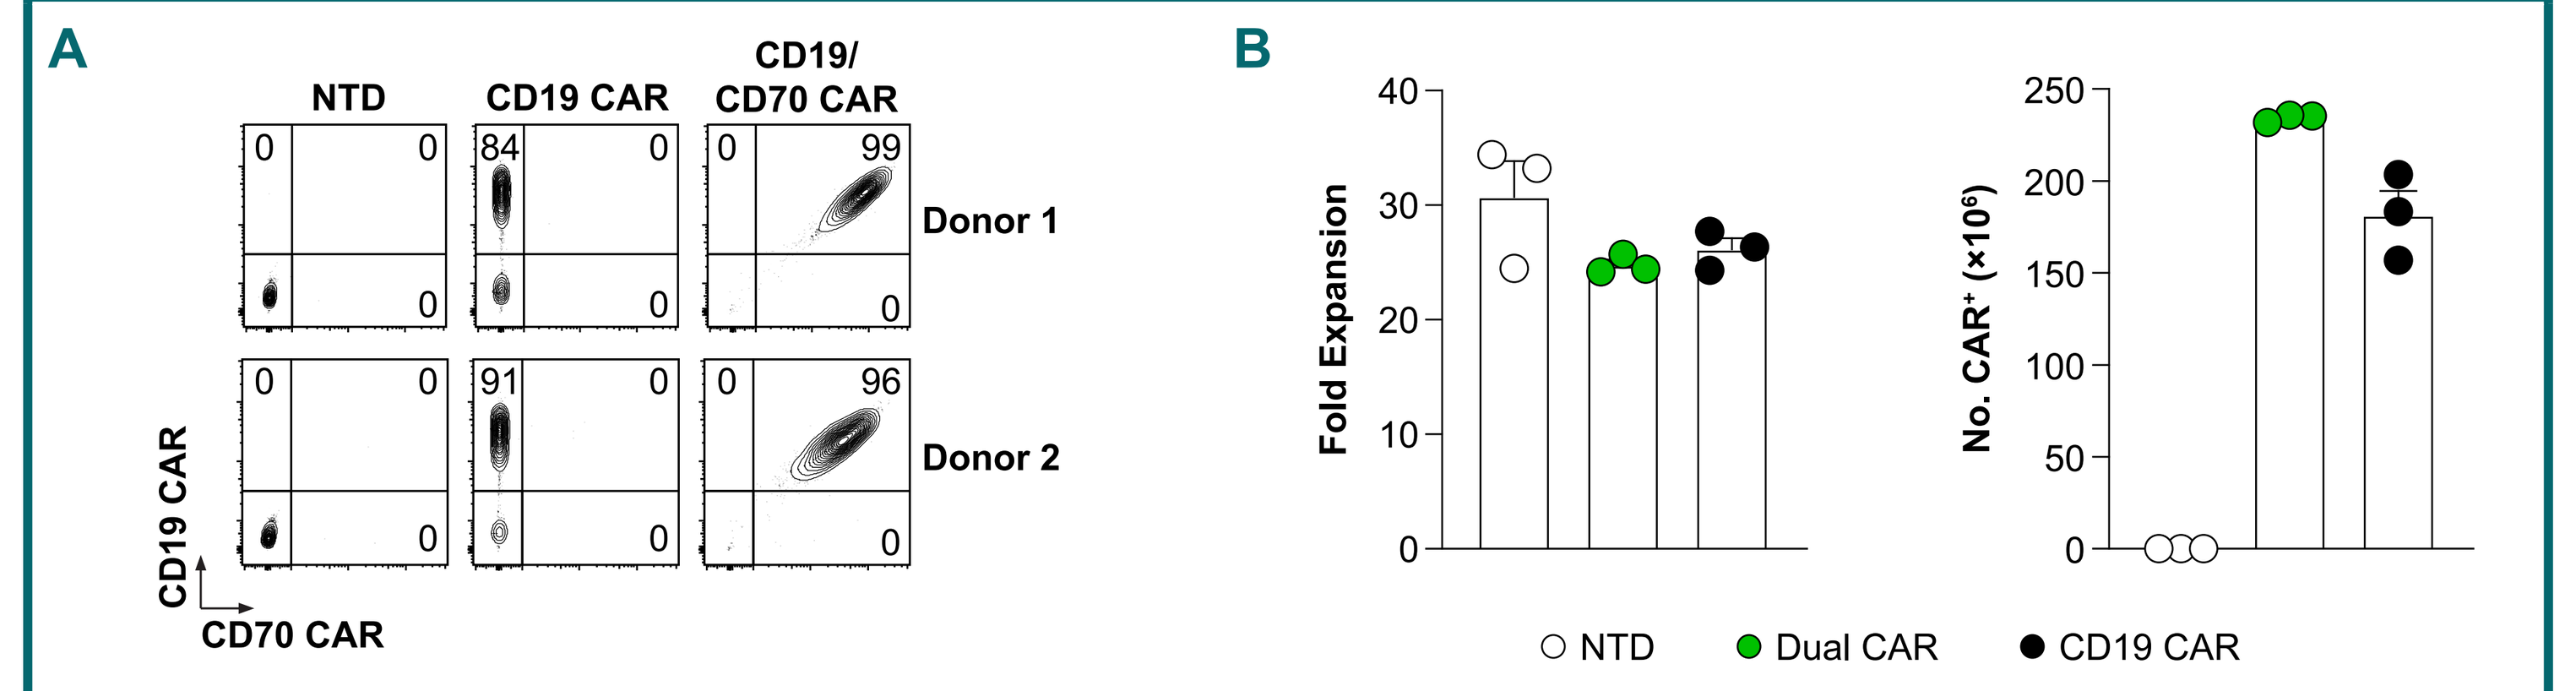
A
NTD
CD19 CAR
CD19/ CD70 CAR
Donor 1
Donor 2
CD19 CAR
CD70 CAR
B
Fold Expansion
No. CAR⁺ (×10⁶)
NTD
Dual CAR
CD19 CAR

Following expansion, dual CAR T cells were characterized by flow cytometry analysis of CAR expression at the end of production (day 14). (A) Representative flow cytometry plots showing CD19 CAR and CD70 CAR expression from 2 donors. (B) Fold expansion of CAR T cells and total number of CAR⁺ cells in the final product. Symbols represent individual donors.
CAR, chimeric antigen receptor; NTD, non-transduced T cells.

Figure 3. ALLO-329 CAR T Cells Exhibit Dual Targeting

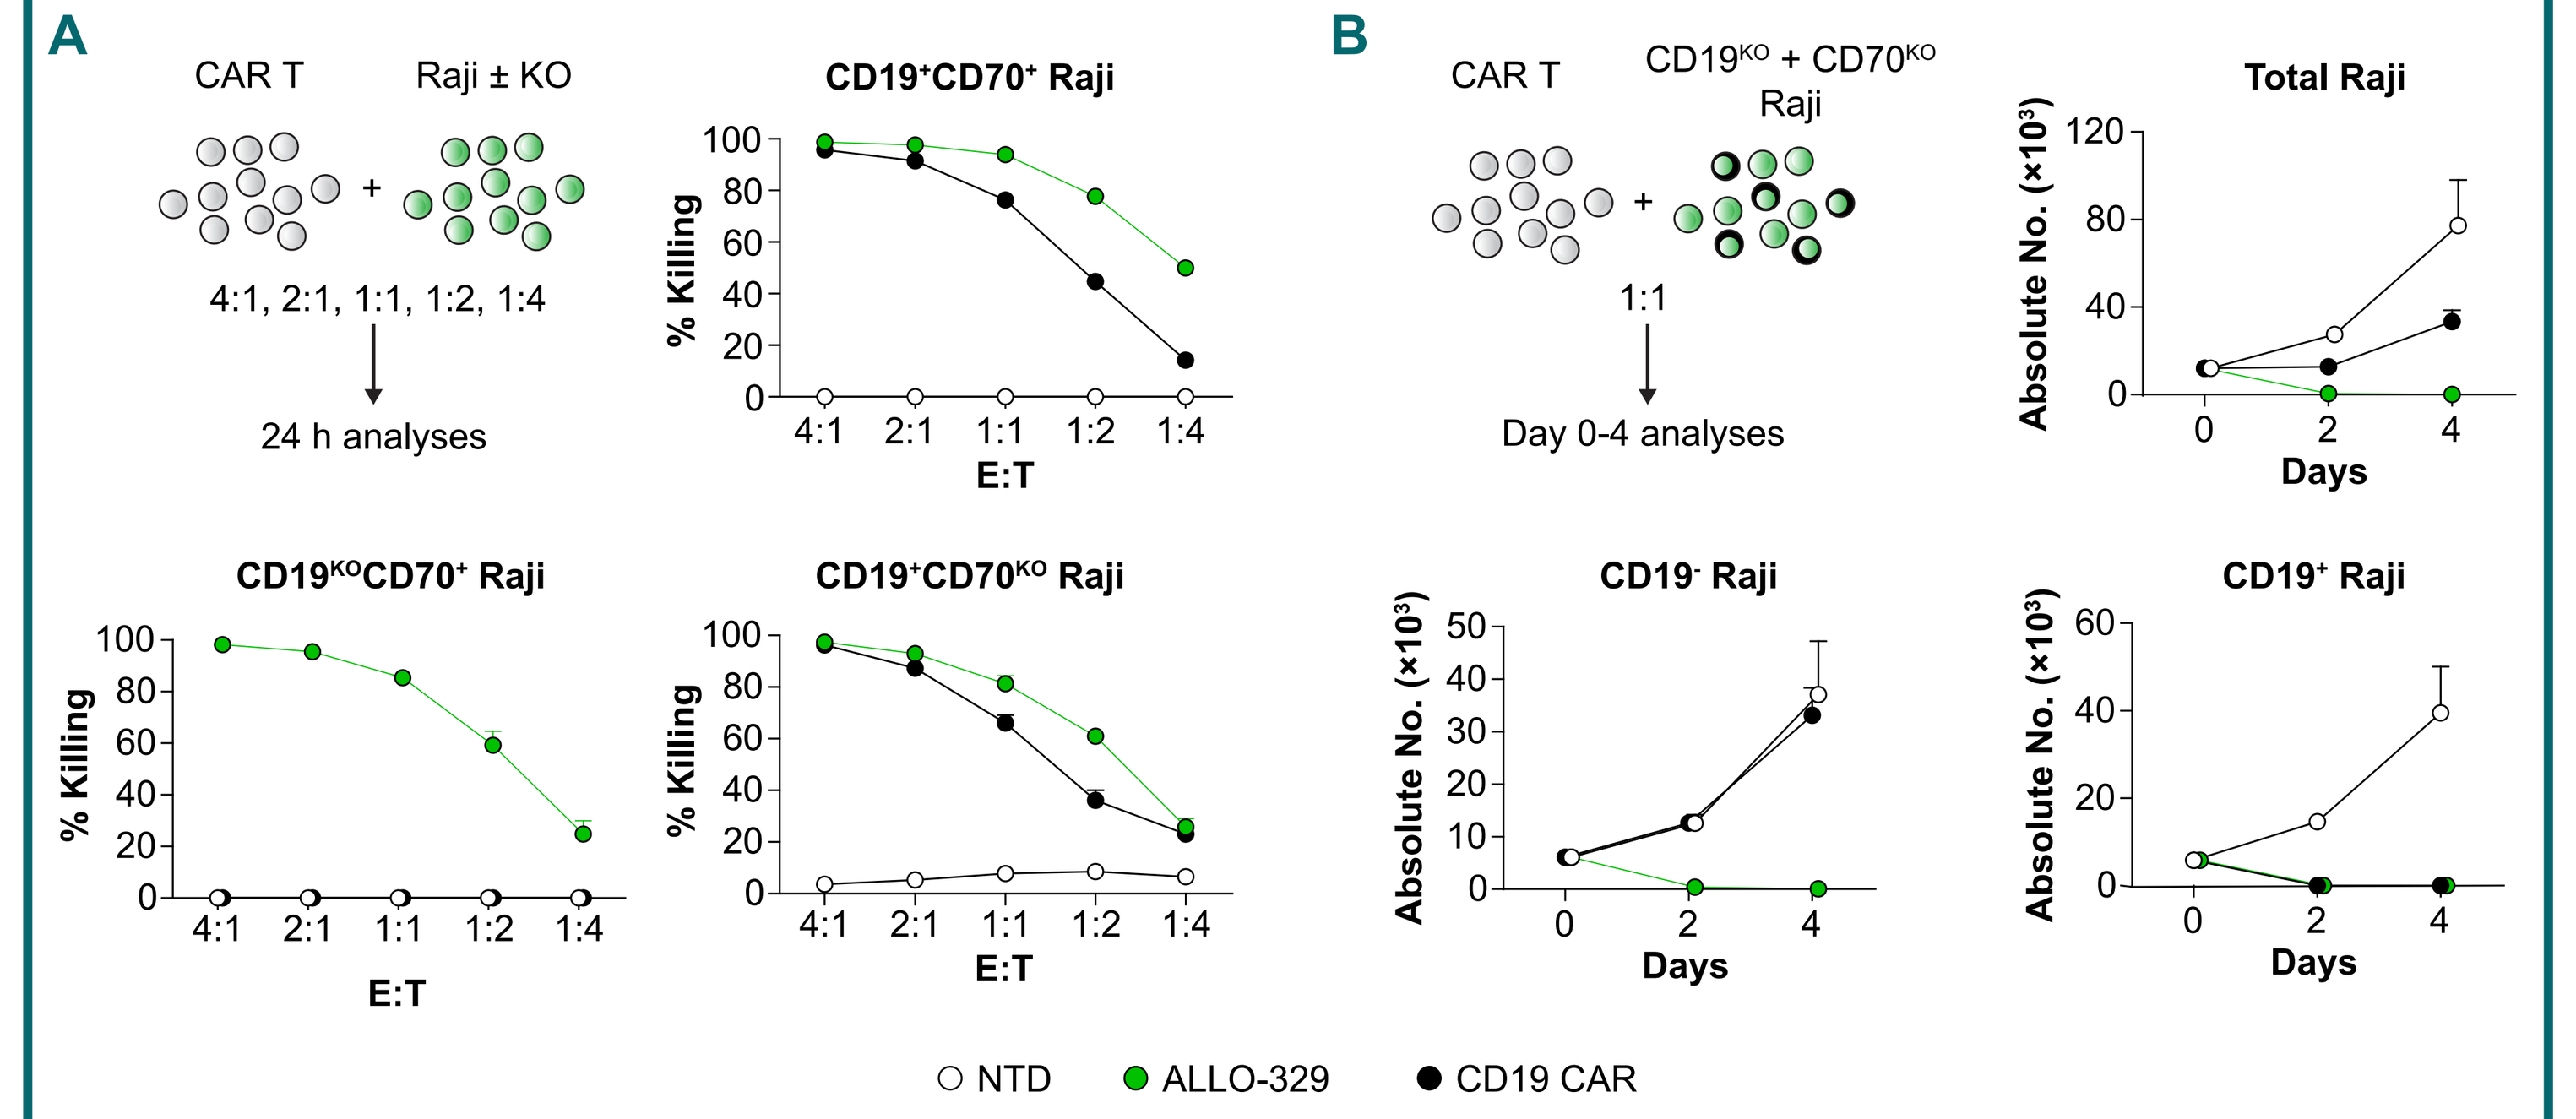
A
CAR T
Raji ± KO
4:1, 2:1, 1:1, 1:2, 1:4
24 h analyses
CD19⁺CD70⁺ Raji
CD19KOCD70⁺ Raji
CD19⁺CD70KO Raji
% Killing
E:T
B
CAR T
CD19KO + CD70KO Raji
1:1
Day 0-4 analyses
Total Raji
CD19⁻ Raji
CD19⁺ Raji
Absolute No. (×10³)
Days
NTD
ALLO-329
CD19 CAR

Cytotoxicity and dual targeting activity of ALLO-329 were assessed *in vitro* using Raji cell lines expressing GFP and luciferase. (A) 24 h short-term killing assay with luciferase-based readout. (B) CD19KO and CD70KO Raji cells were mixed at a ratio of 1:1. E:T ratios of 1:1 were tested using mixed tumor targets. Flow cytometry-based analysis was performed at the indicated times.
CAR, chimeric antigen receptor; E:T, effector to target; GFP, green fluorescent protein; KO, knockout; NTD, non-transduced T cells.

Figure 4. ALLO-329 CAR T Cells Resist Rejection by Alloreactive T Cells From SLE Donors

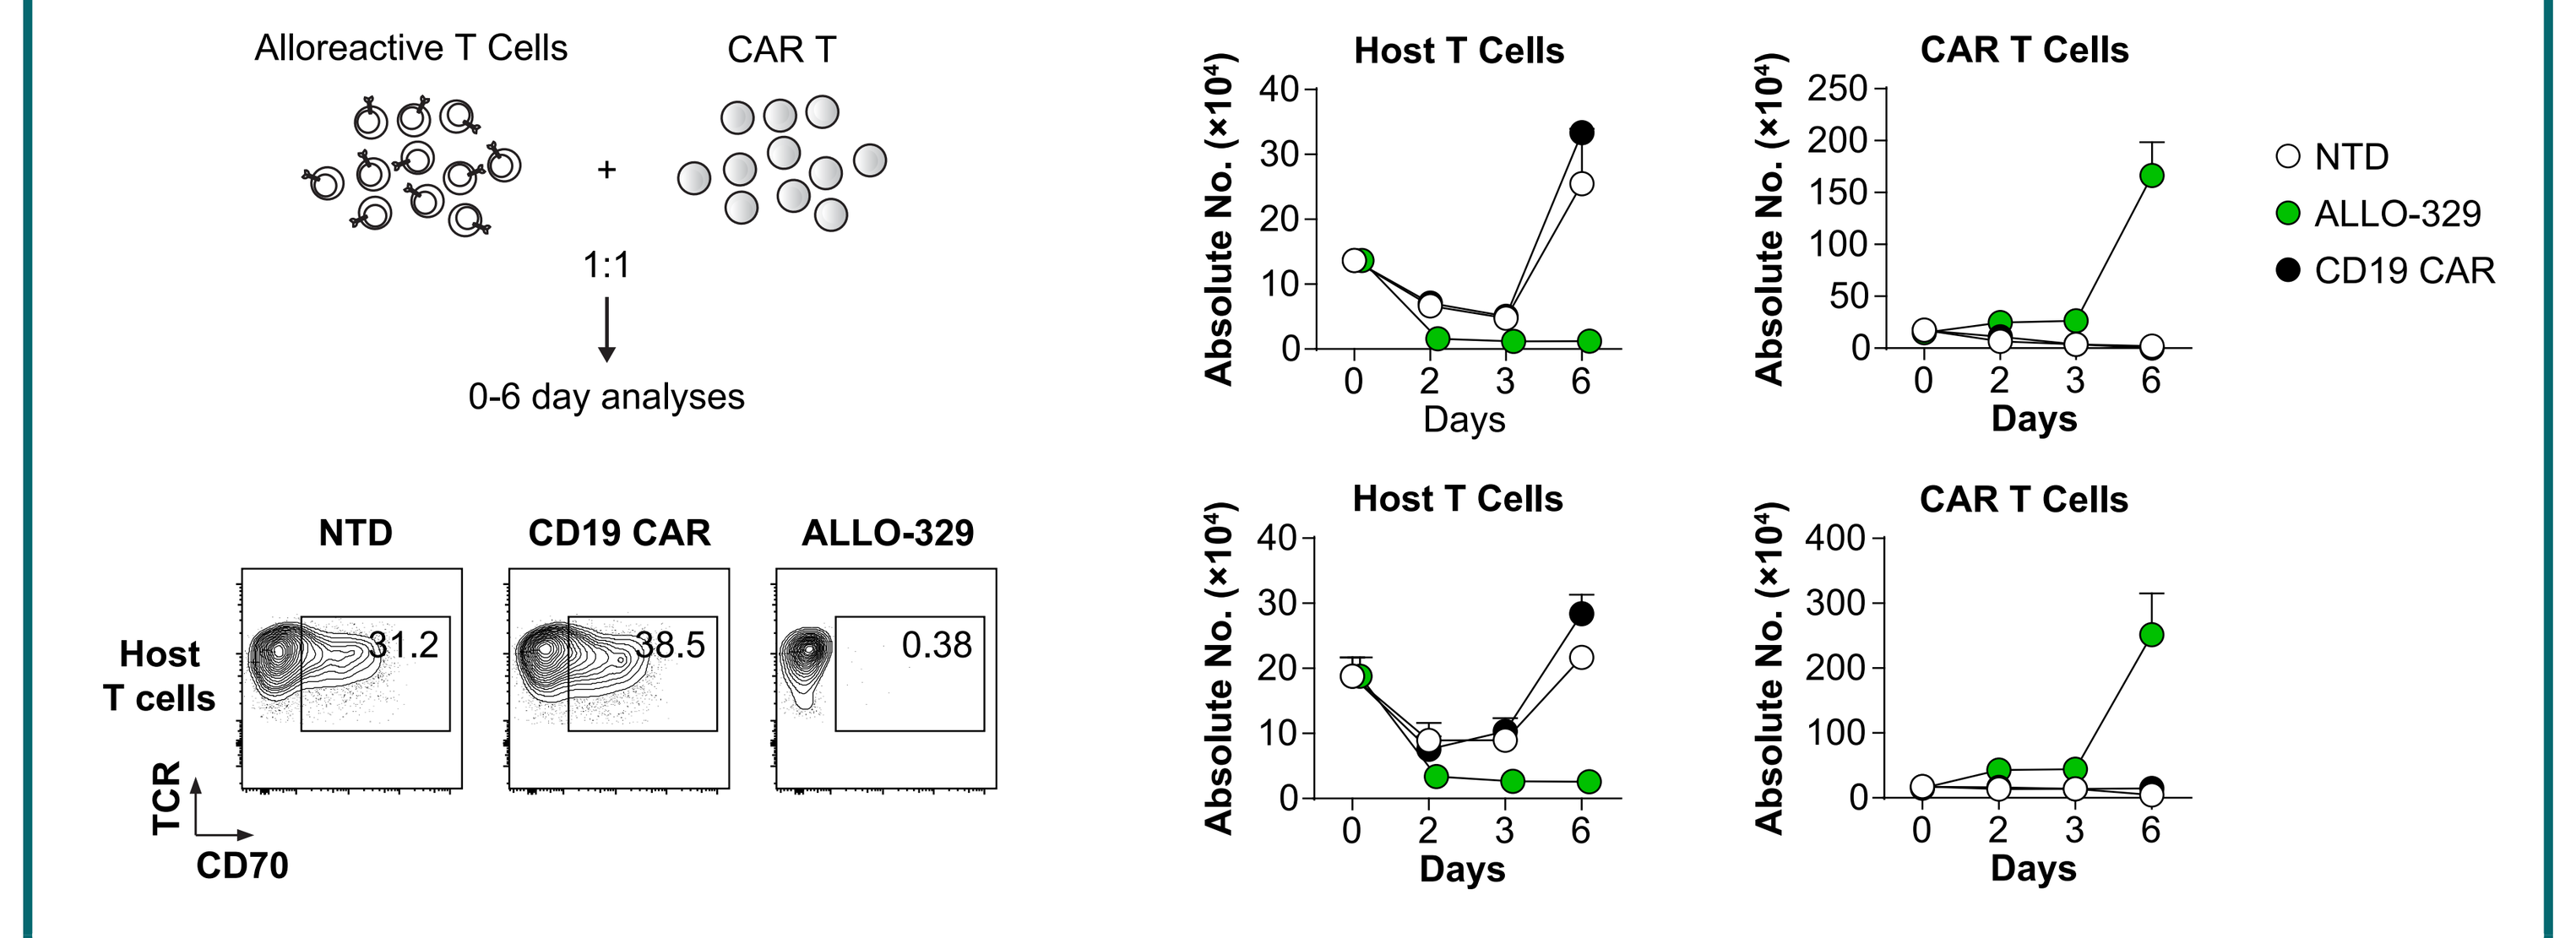
Alloreactive T Cells
CAR T
1:1
0-6 day analyses
NTD
CD19 CAR
ALLO-329
Host T cells
31.2
38.5
0.38
TCR
CD70
Host T Cells
CAR T Cells
Absolute No. (×10⁴)
Days
NTD
ALLO-329
CD19 CAR

The susceptibility of ALLO-329 to rejection by HLA-mismatched T cells was assessed in MLR assays. A total of 4 unique graft:host donor pairs were tested (2 healthy and 2 SLE pairs). Data are the average of technical replicates. Representative graft:host donor pair is shown. Counts are normalized to graft-only controls. Flow cytometry plots show representative data from day 6. Healthy donor (top row). SLE donor (bottom row).
CAR, chimeric antigen receptor; ctrl, control; HLA, human leukocyte antigen; KO, knockout; MLR, mixed lymphocyte reaction; NTD, non-transduced T cells; SLE, systemic lupus erythematosus; TCR, T cell receptor.

Figure 5. B Cells From SLE Donors Are Eliminated by ALLO-329 CAR T Cells *In Vitro*

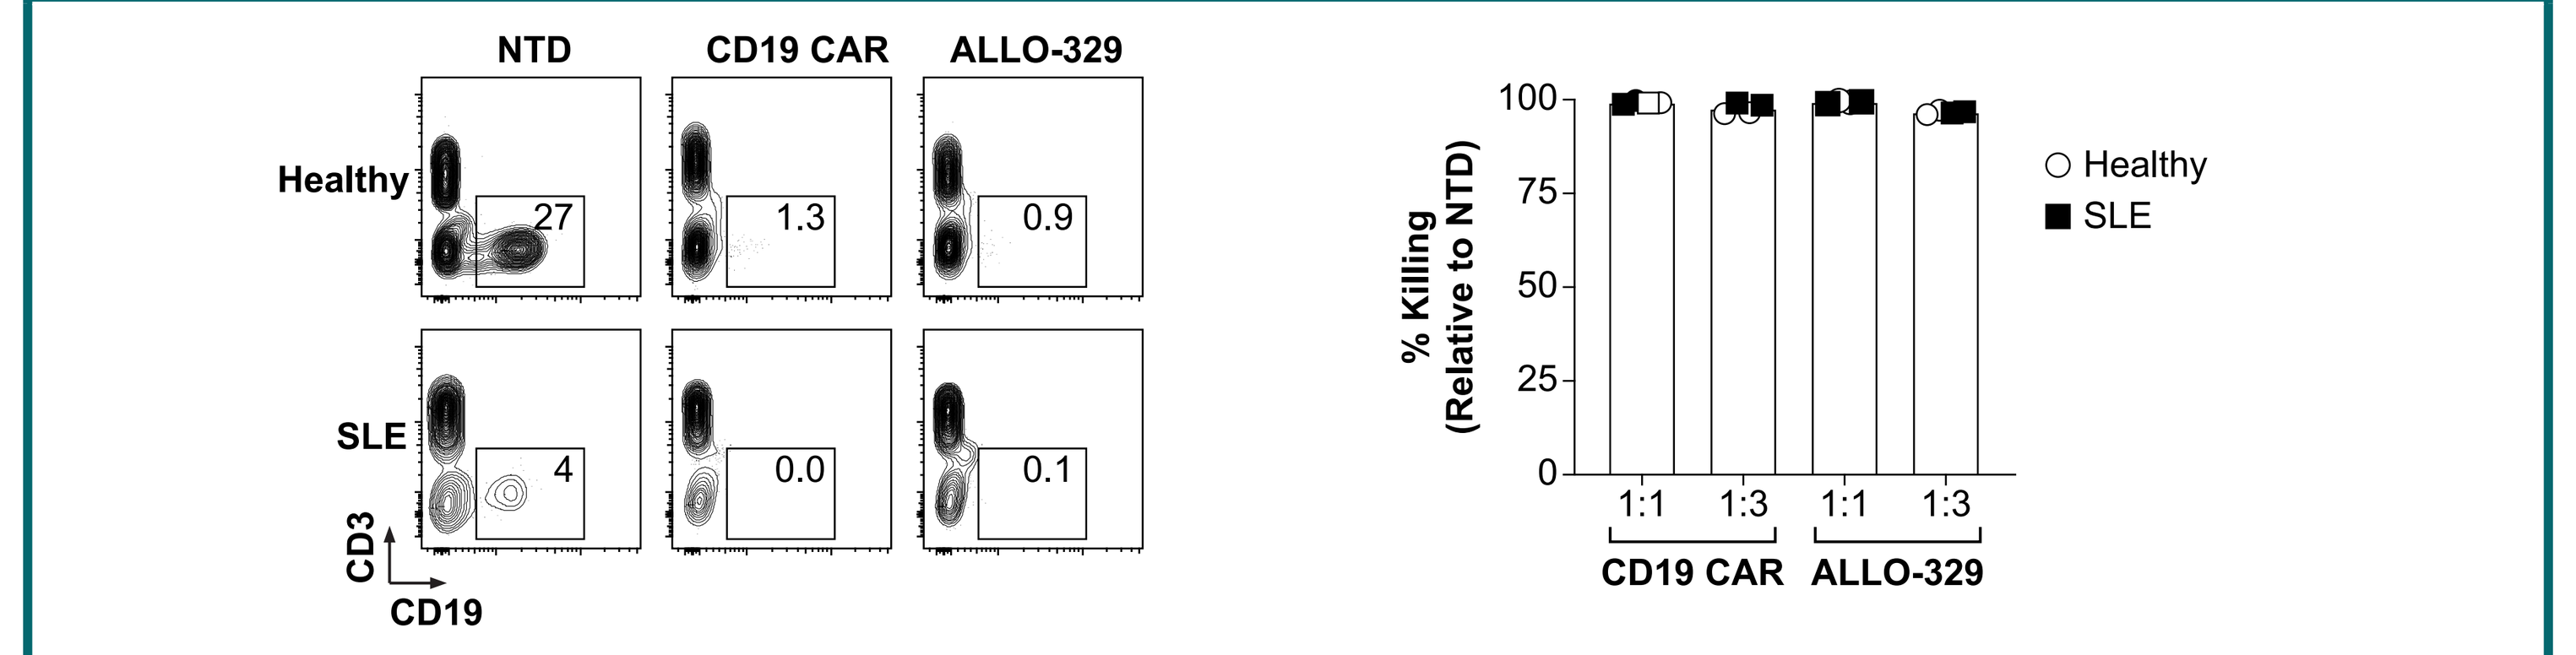
NTD
CD19 CAR
ALLO-329
Healthy
SLE
27
1.3
0.9
4
0.0
0.1
CD3
CD19
% Killing (Relative to NTD)
1:1
1:3
CD19 CAR
ALLO-329
Healthy
SLE

PBMCs from healthy or SLE donors were co-cultured with CAR T cells at an E:T ratio of 1:1 or 1:3 for 24 h. E:T ratio of 1:3 shown for flow cytometry plots; effector cells gated out. Read out was determined by flow cytometry and is plotted relative to NTD control. Symbols represent unique host/graft donor pairs.
CAR, chimeric antigen receptor; E:T, effector to target; NTD, non-transduced T cells; PBMC, peripheral blood mononuclear cell; SLE, systemic lupus erythematosus.

Figure 6. Dose-Dependent Responses to ALLO-329 Treatment in a Mouse Model of B Cell Lymphoma

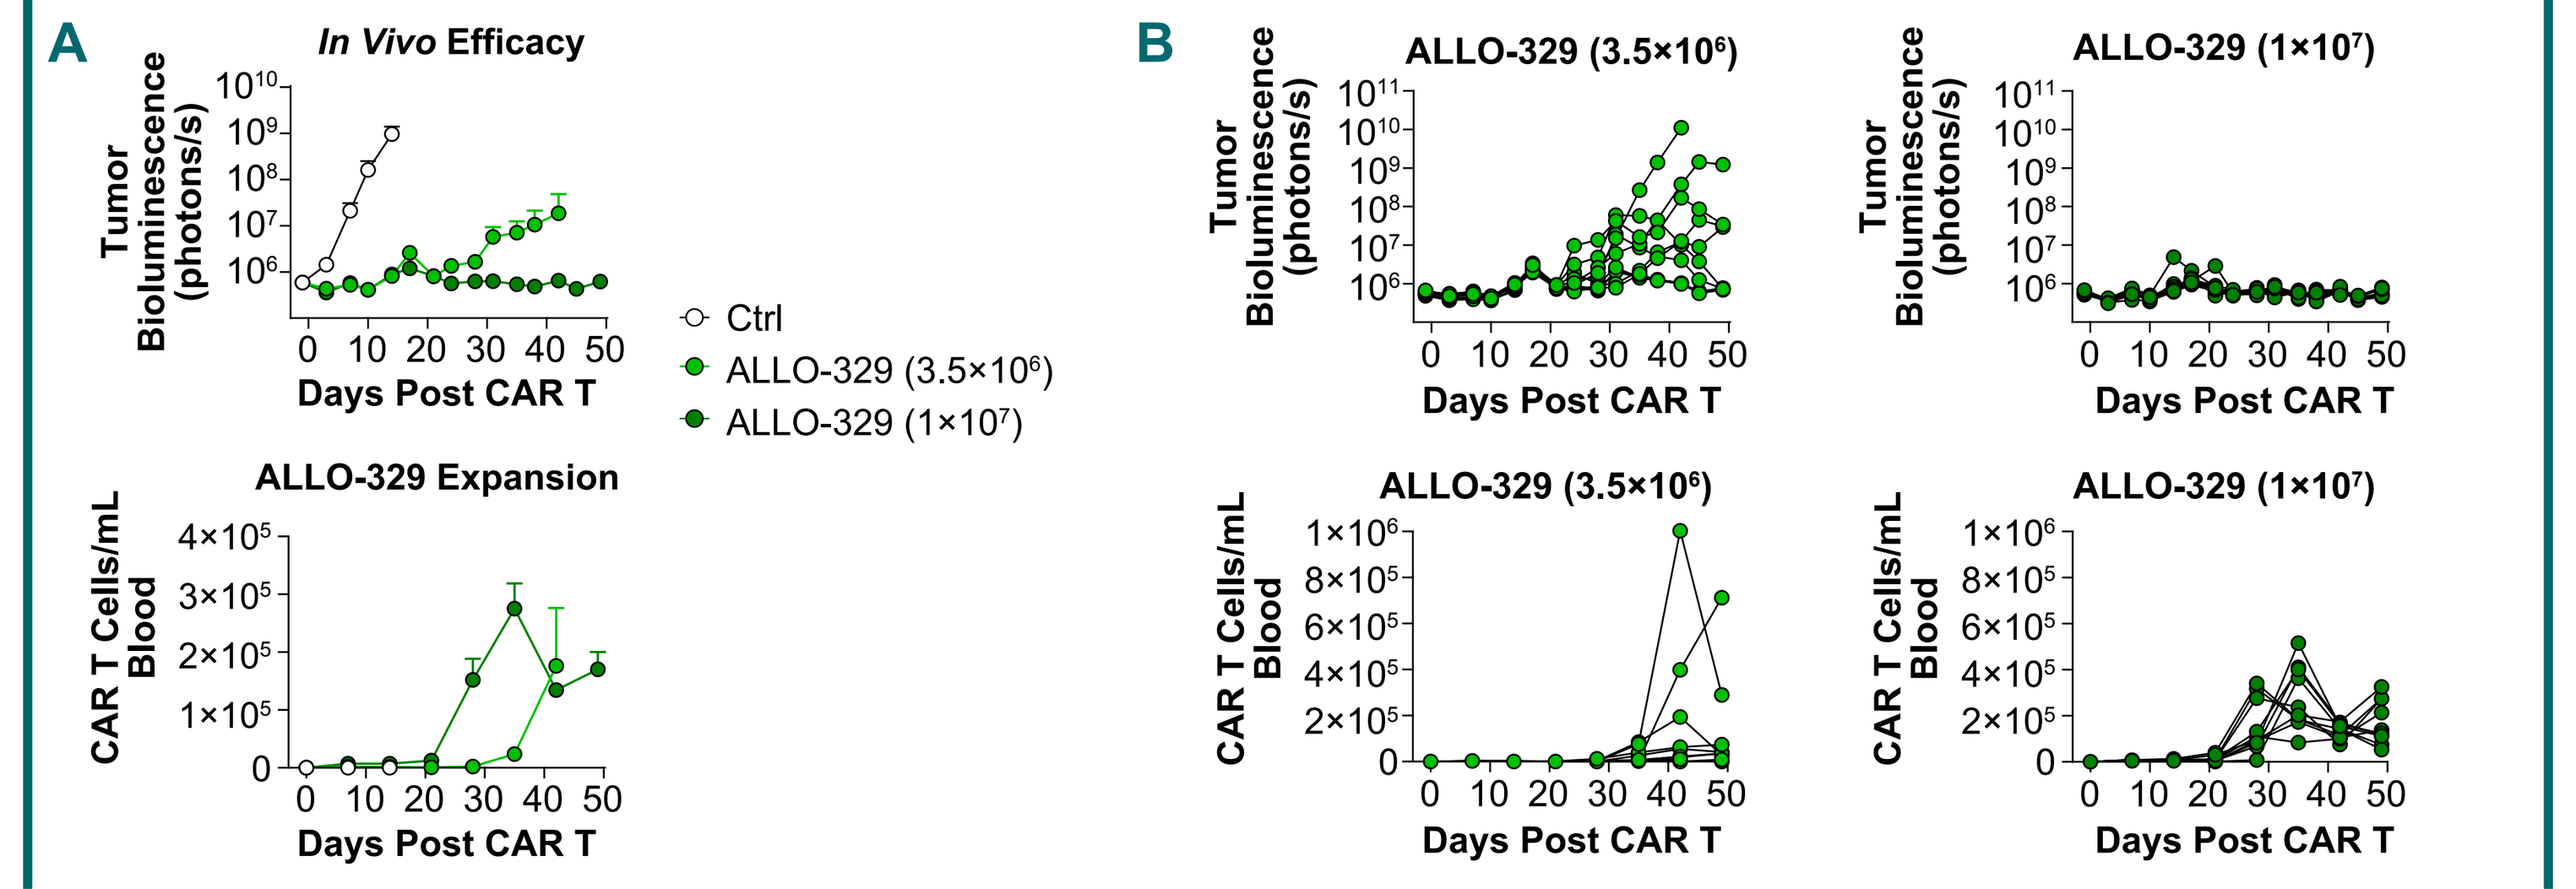
A
In Vivo Efficacy
Tumor Bioluminescence (photons/s)
Days Post CAR T
Ctrl
ALLO-329 (3.5×10⁶)
ALLO-329 (1×10⁷)
ALLO-329 Expansion
CAR T Cells/mL Blood
B
ALLO-329 (3.5×10⁶)
ALLO-329 (1×10⁷)

In vivo study performed using a B cell lymphoma model. Wild-type Raji cells (5×10⁵) were injected into NSG mice 4 days prior to ALLO-329 treatment (n=10). (A) Data pooled. (B) Individual mice data.
CAR, chimeric antigen receptor; ctrl, control; NSG, NOD-*scid* IL2Rgamma^null^.

Figure 7. Rapid B Cell Depletion and Reduction in Immunoglobulin Levels in Mice Receiving ALLO-329 CAR T Cells

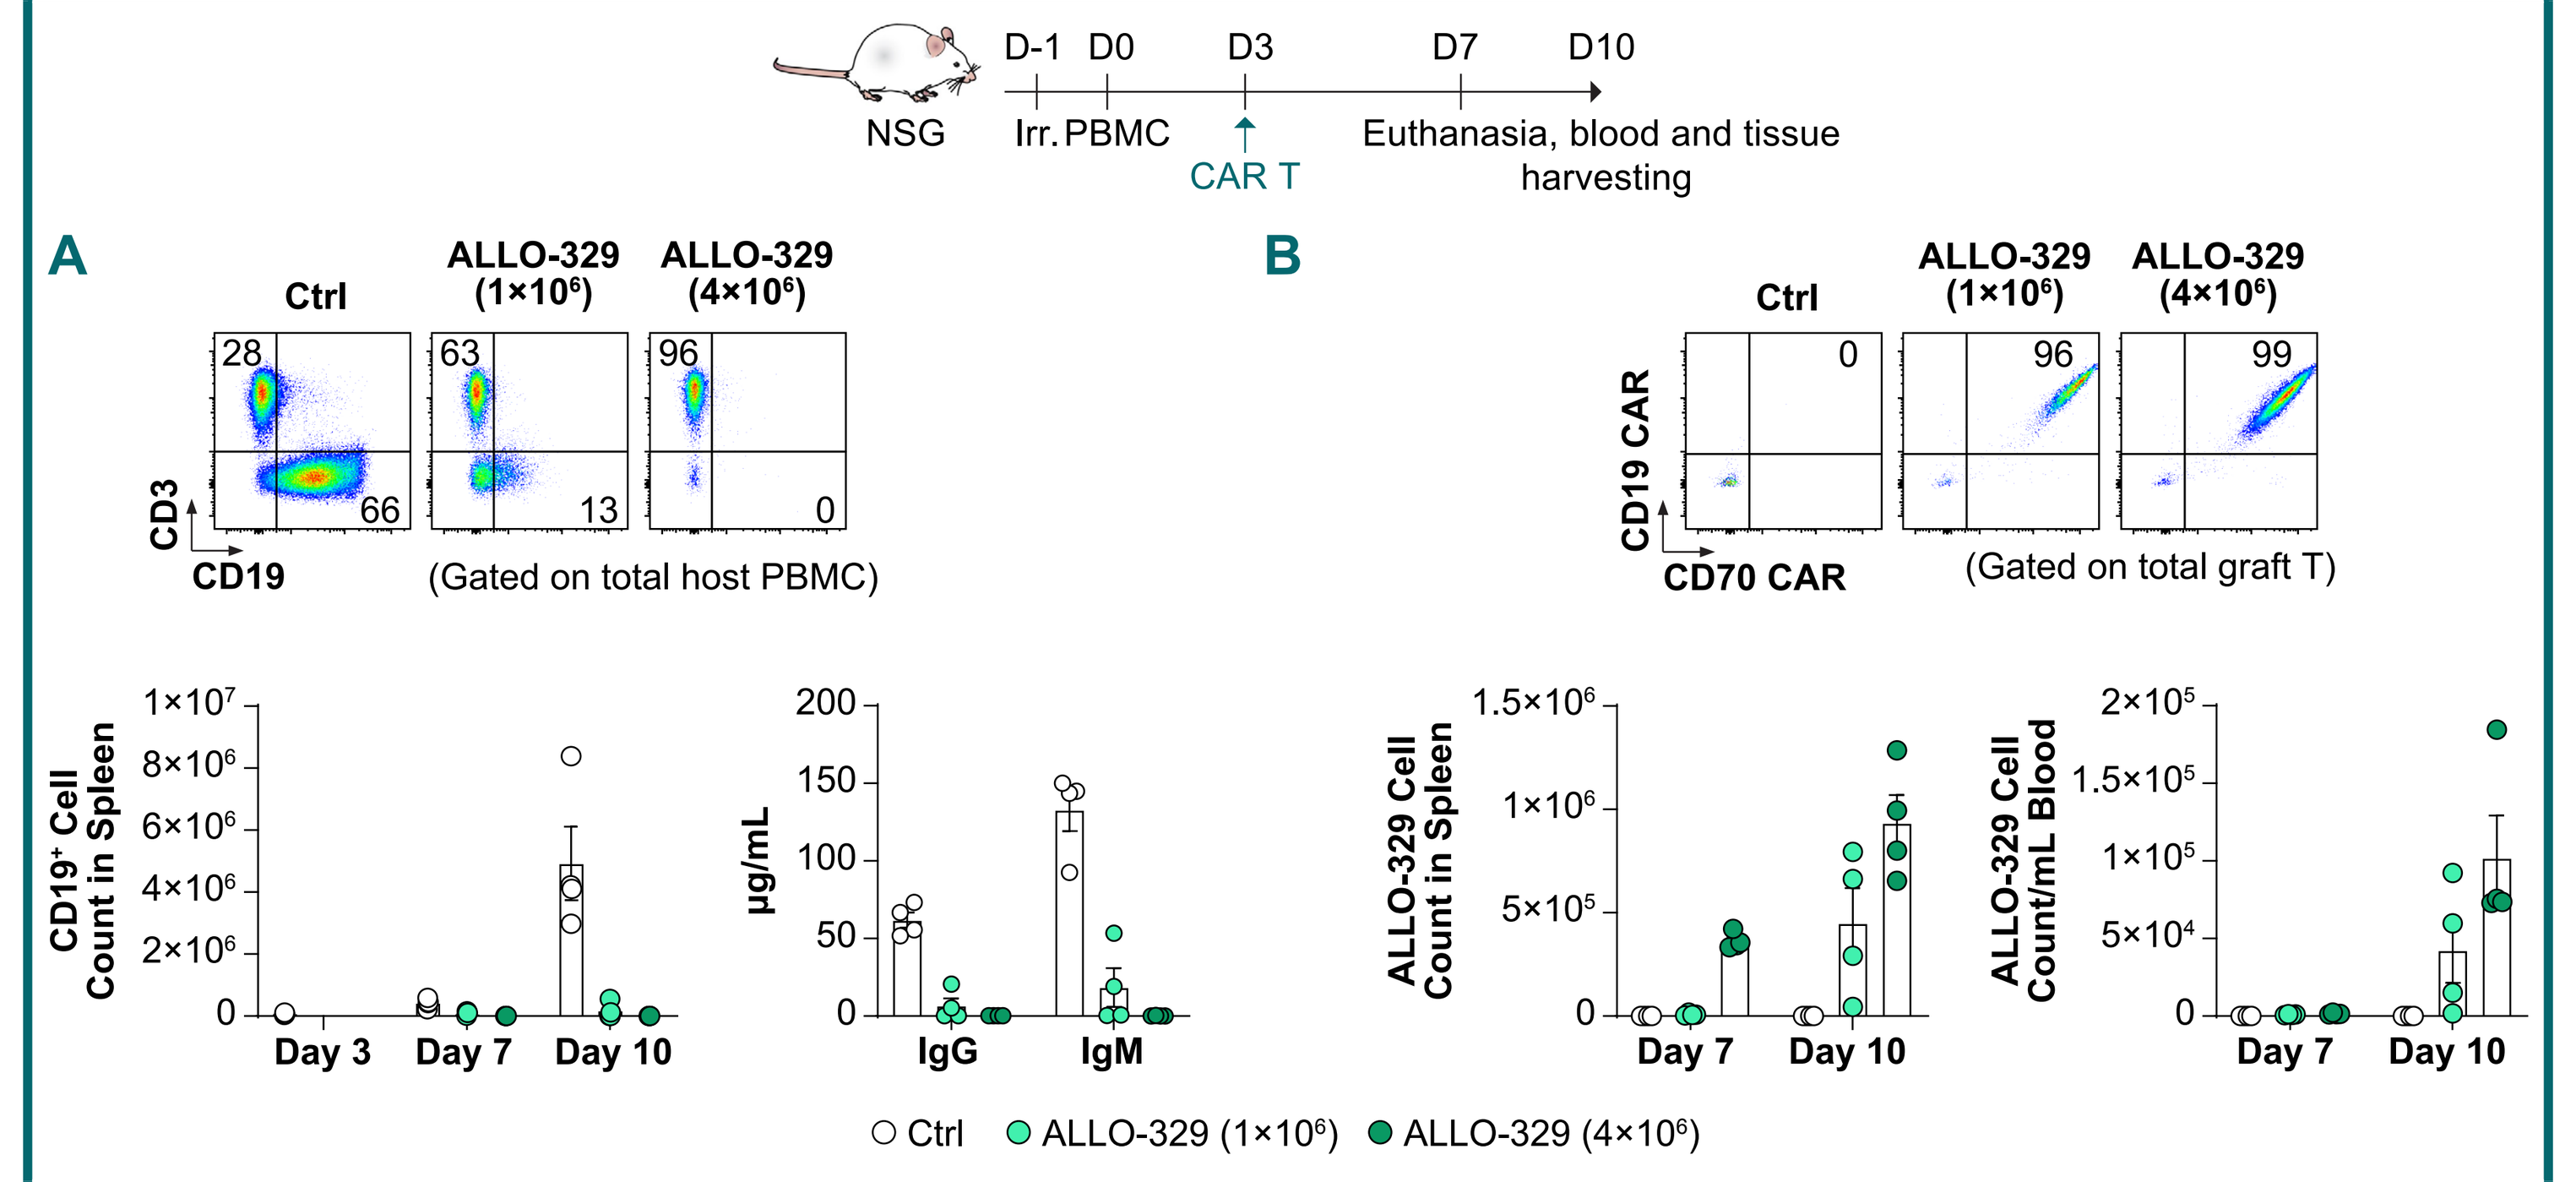
NSG
D-1
D0
D3
D7
D10
Irr. PBMC
CAR T
Euthanasia, blood and tissue harvesting
A
Ctrl
ALLO-329 (1×10⁶)
ALLO-329 (4×10⁶)
CD3
CD19
(Gated on total host PBMC)
B
CD19 CAR
CD70 CAR
(Gated on total graft T)
CD19⁺ Cell Count in Spleen
µg/mL
IgG
IgM
ALLO-329 Cell Count in Spleen
ALLO-329 Cell Count/mL Blood
Day 3
Day 7
Day 10
Ctrl
ALLO-329 (1×10⁶)
ALLO-329 (4×10⁶)

Two healthy donor PBMCs were tested (2×10⁷ cells/mouse); data from representative donors are shown. Four NSG mice/group were evaluated. Mice received 1 Gy irradiation on day -1. Graft and host population are distinguished by HLA-A2. Day 10 flow cytometry plots are shown. Levels of human antibodies in the plasma were measured on day 10.
CAR, chimeric antigen receptor; ctrl, control; D, day; irr, irradiation; HLA, human leukocyte antigen; NSG, NOD-*scid* IL2Rgamma^null^; PBMC, peripheral blood mononuclear cell.

Figure 8. ALLO-329 Eliminates CD70⁺ T Cells in Mice Engrafted With Human PBMCs

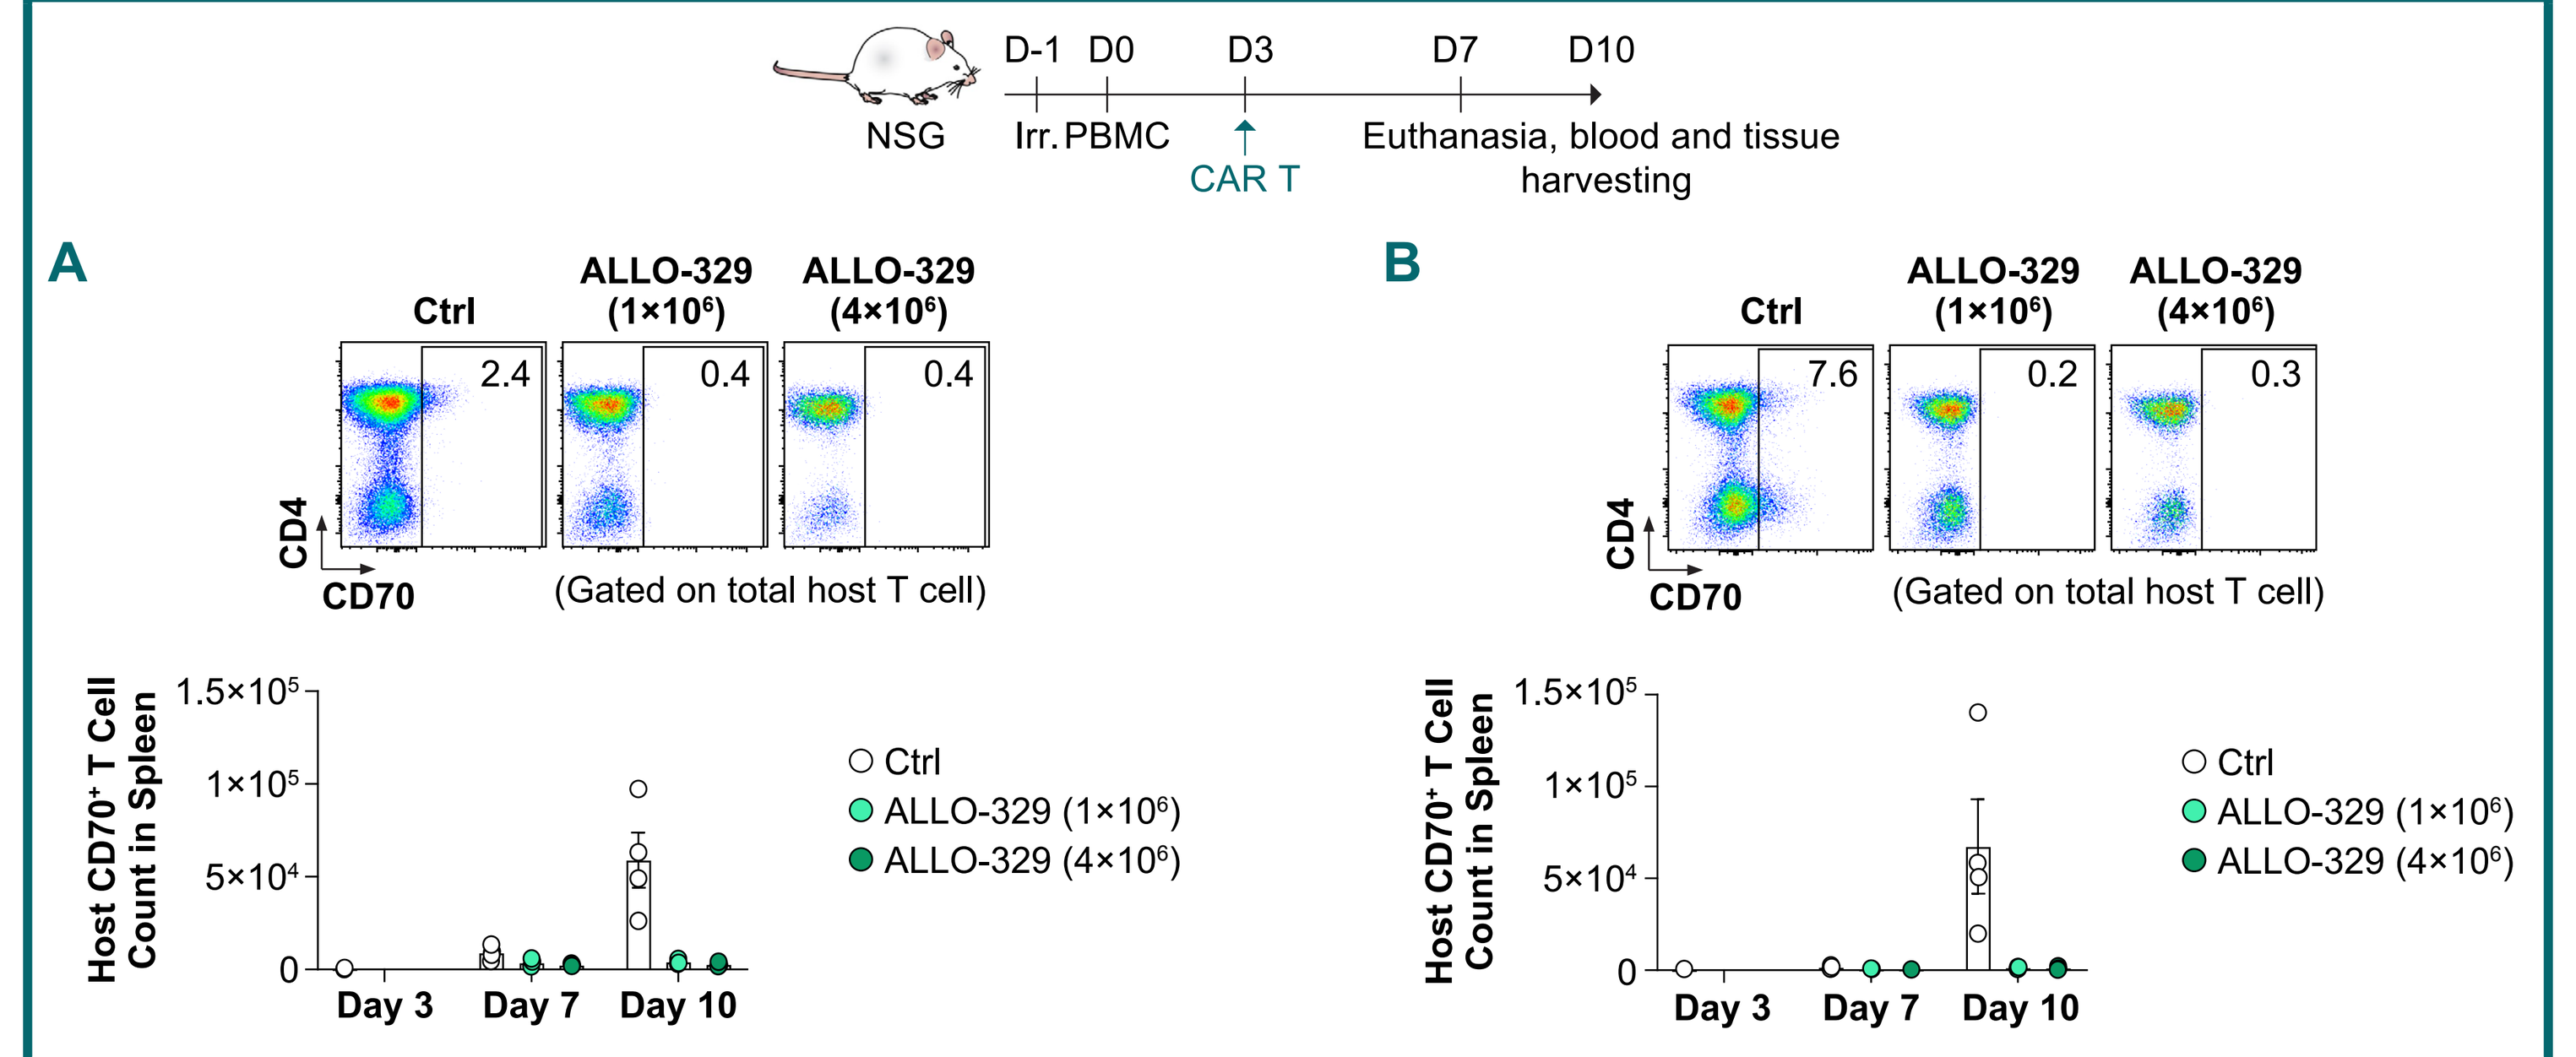
NSG
D-1
D0
D3
D7
D10
Irr. PBMC
CAR T
Euthanasia, blood and tissue harvesting
A
B
Ctrl
ALLO-329 (1×10⁶)
ALLO-329 (4×10⁶)
CD4
CD70
(Gated on total host T cell)
Host CD70⁺ T Cell Count in Spleen
Day 3
Day 7
Day 10

Two healthy donor PBMCs were tested (2×10⁷ cells/mouse); data from representative donors are shown. Four NSG mice/group were evaluated. Mice received 1 Gy irradiation on day -1. Graft and host population are distinguished by HLA-A2. Day 10 flow cytometry plots are shown.
CAR, chimeric antigen receptor; Ctrl, control; D, day; irr, irradiation; HLA, human leukocyte antigen; NSG, NOD-*scid* IL2Rgamma^null^; PBMC, peripheral blood mononuclear cell.

Figure 9. ALLO-329 Transiently Depletes B Cells in Mice Engrafted With Human CD34⁺ HSC

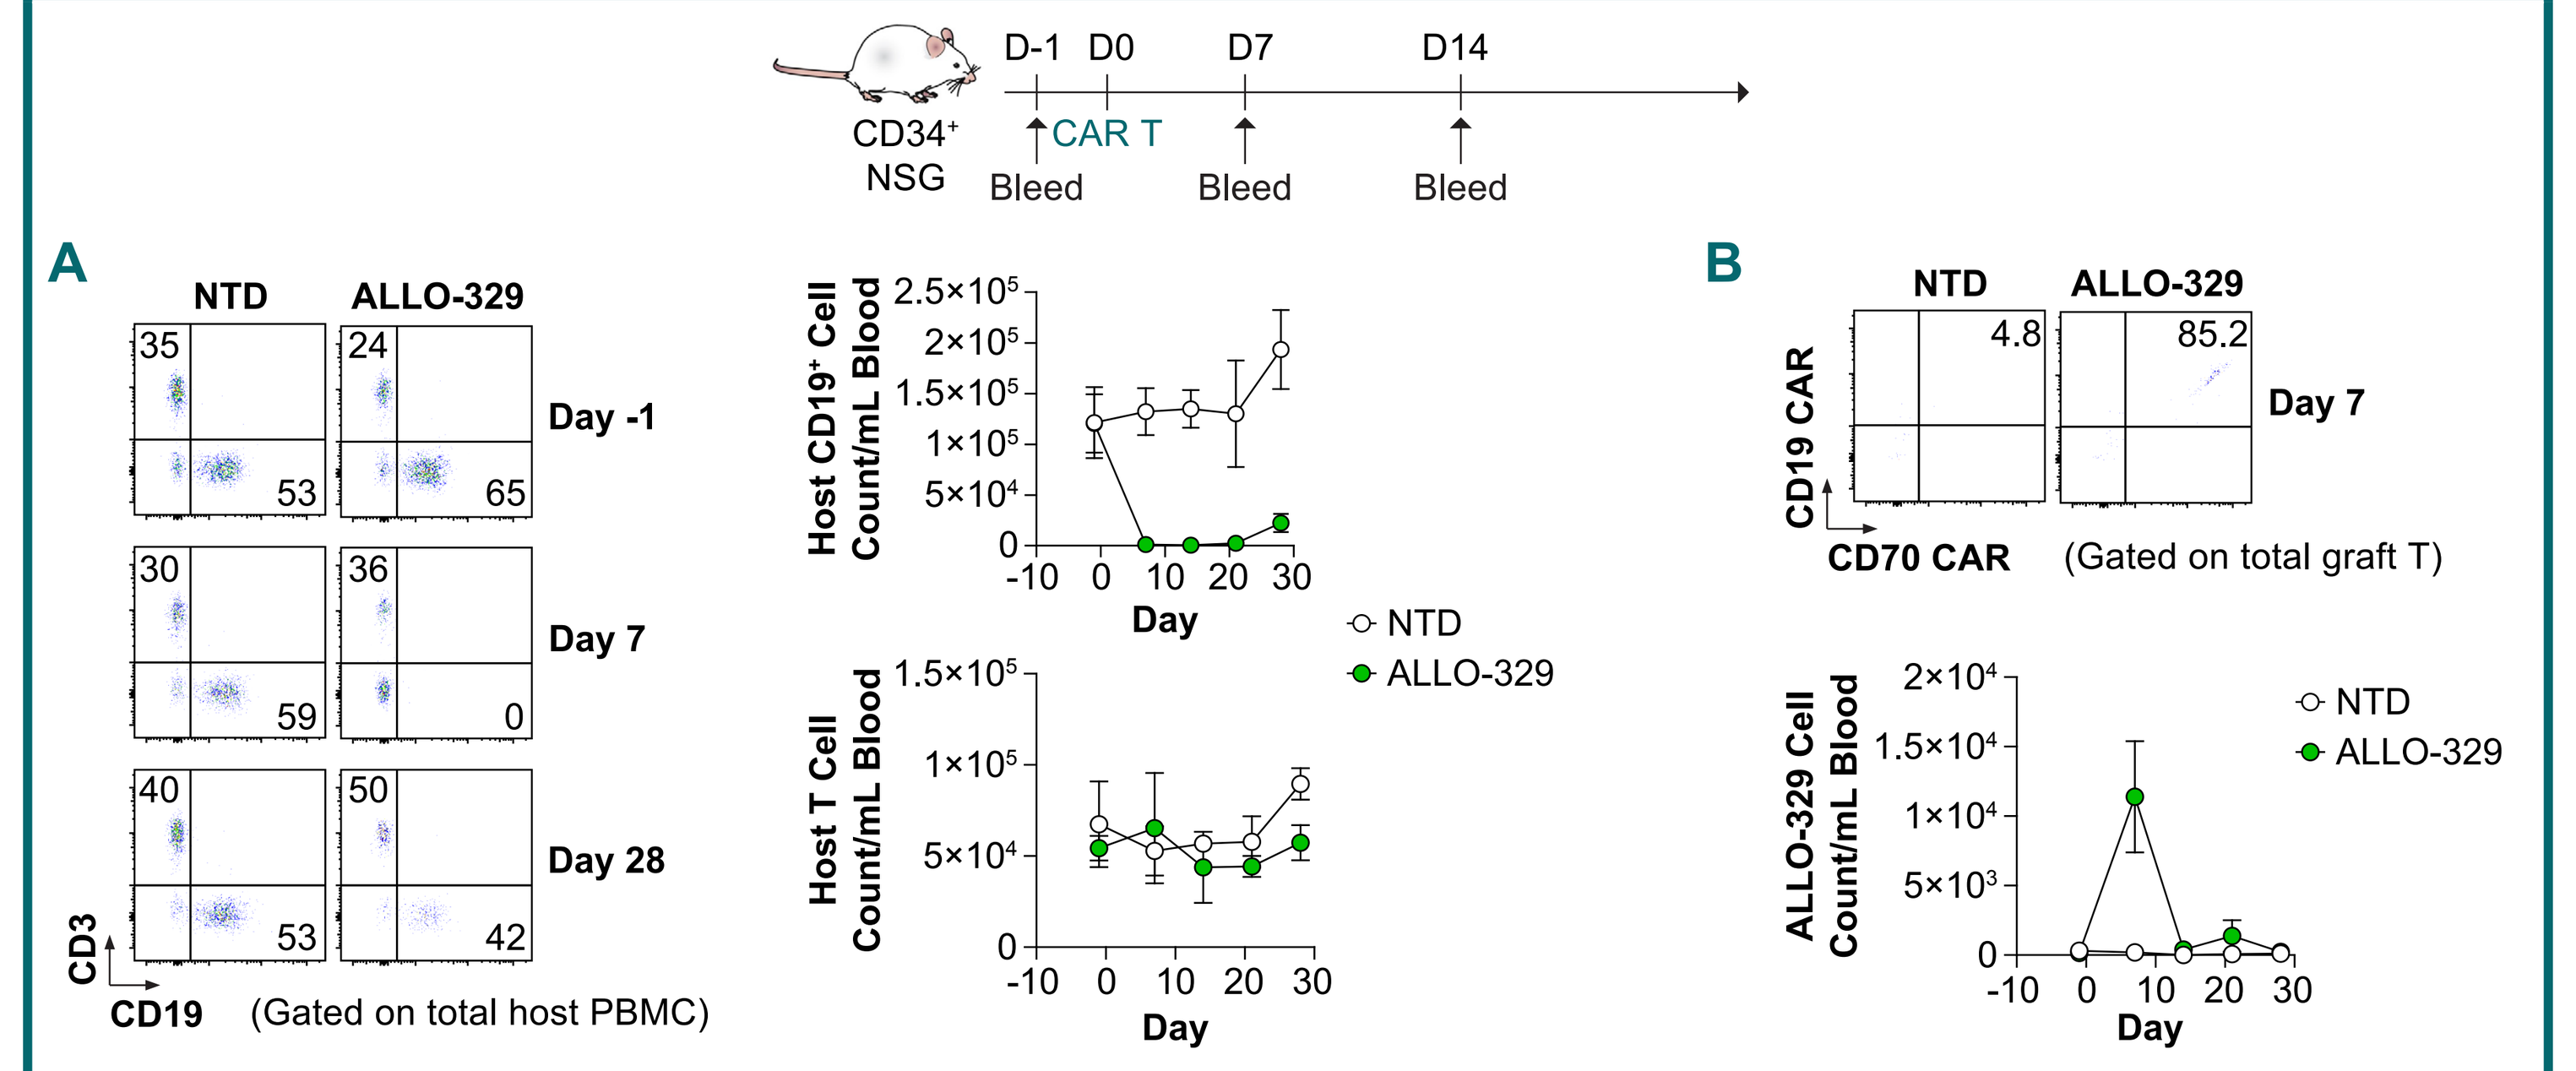
CD34⁺ NSG
D-1
D0
D7
D14
CAR T
Bleed
A
NTD
ALLO-329
Day -1
Day 7
Day 28
CD3
CD19
(Gated on total host PBMC)
Host CD19⁺ Cell Count/mL Blood
Host T Cell Count/mL Blood
Day
NTD
ALLO-329
B
CD19 CAR
CD70 CAR
(Gated on total graft T)
ALLO-329 Cell Count/mL Blood

Three CD34⁺ NSG mice/group were evaluated. Mice were not irradiated or lymphodepleted prior to ALLO-329 treatment (6×10⁶ CAR⁺ cells). Graft and host population are distinguished by HLA-A2.
CAR, chimeric antigen receptor; D, day; HLA, human leukocyte antigen; HSC, hematopoietic stem cell; NSG, NOD-*scid* IL2Rgamma^null^; NTD, non-transduced T cells; PBMC, peripheral blood mononuclear cell.

Figure 10. ALLO-329 Does Not Affect Virus-Specific T Cell Activity

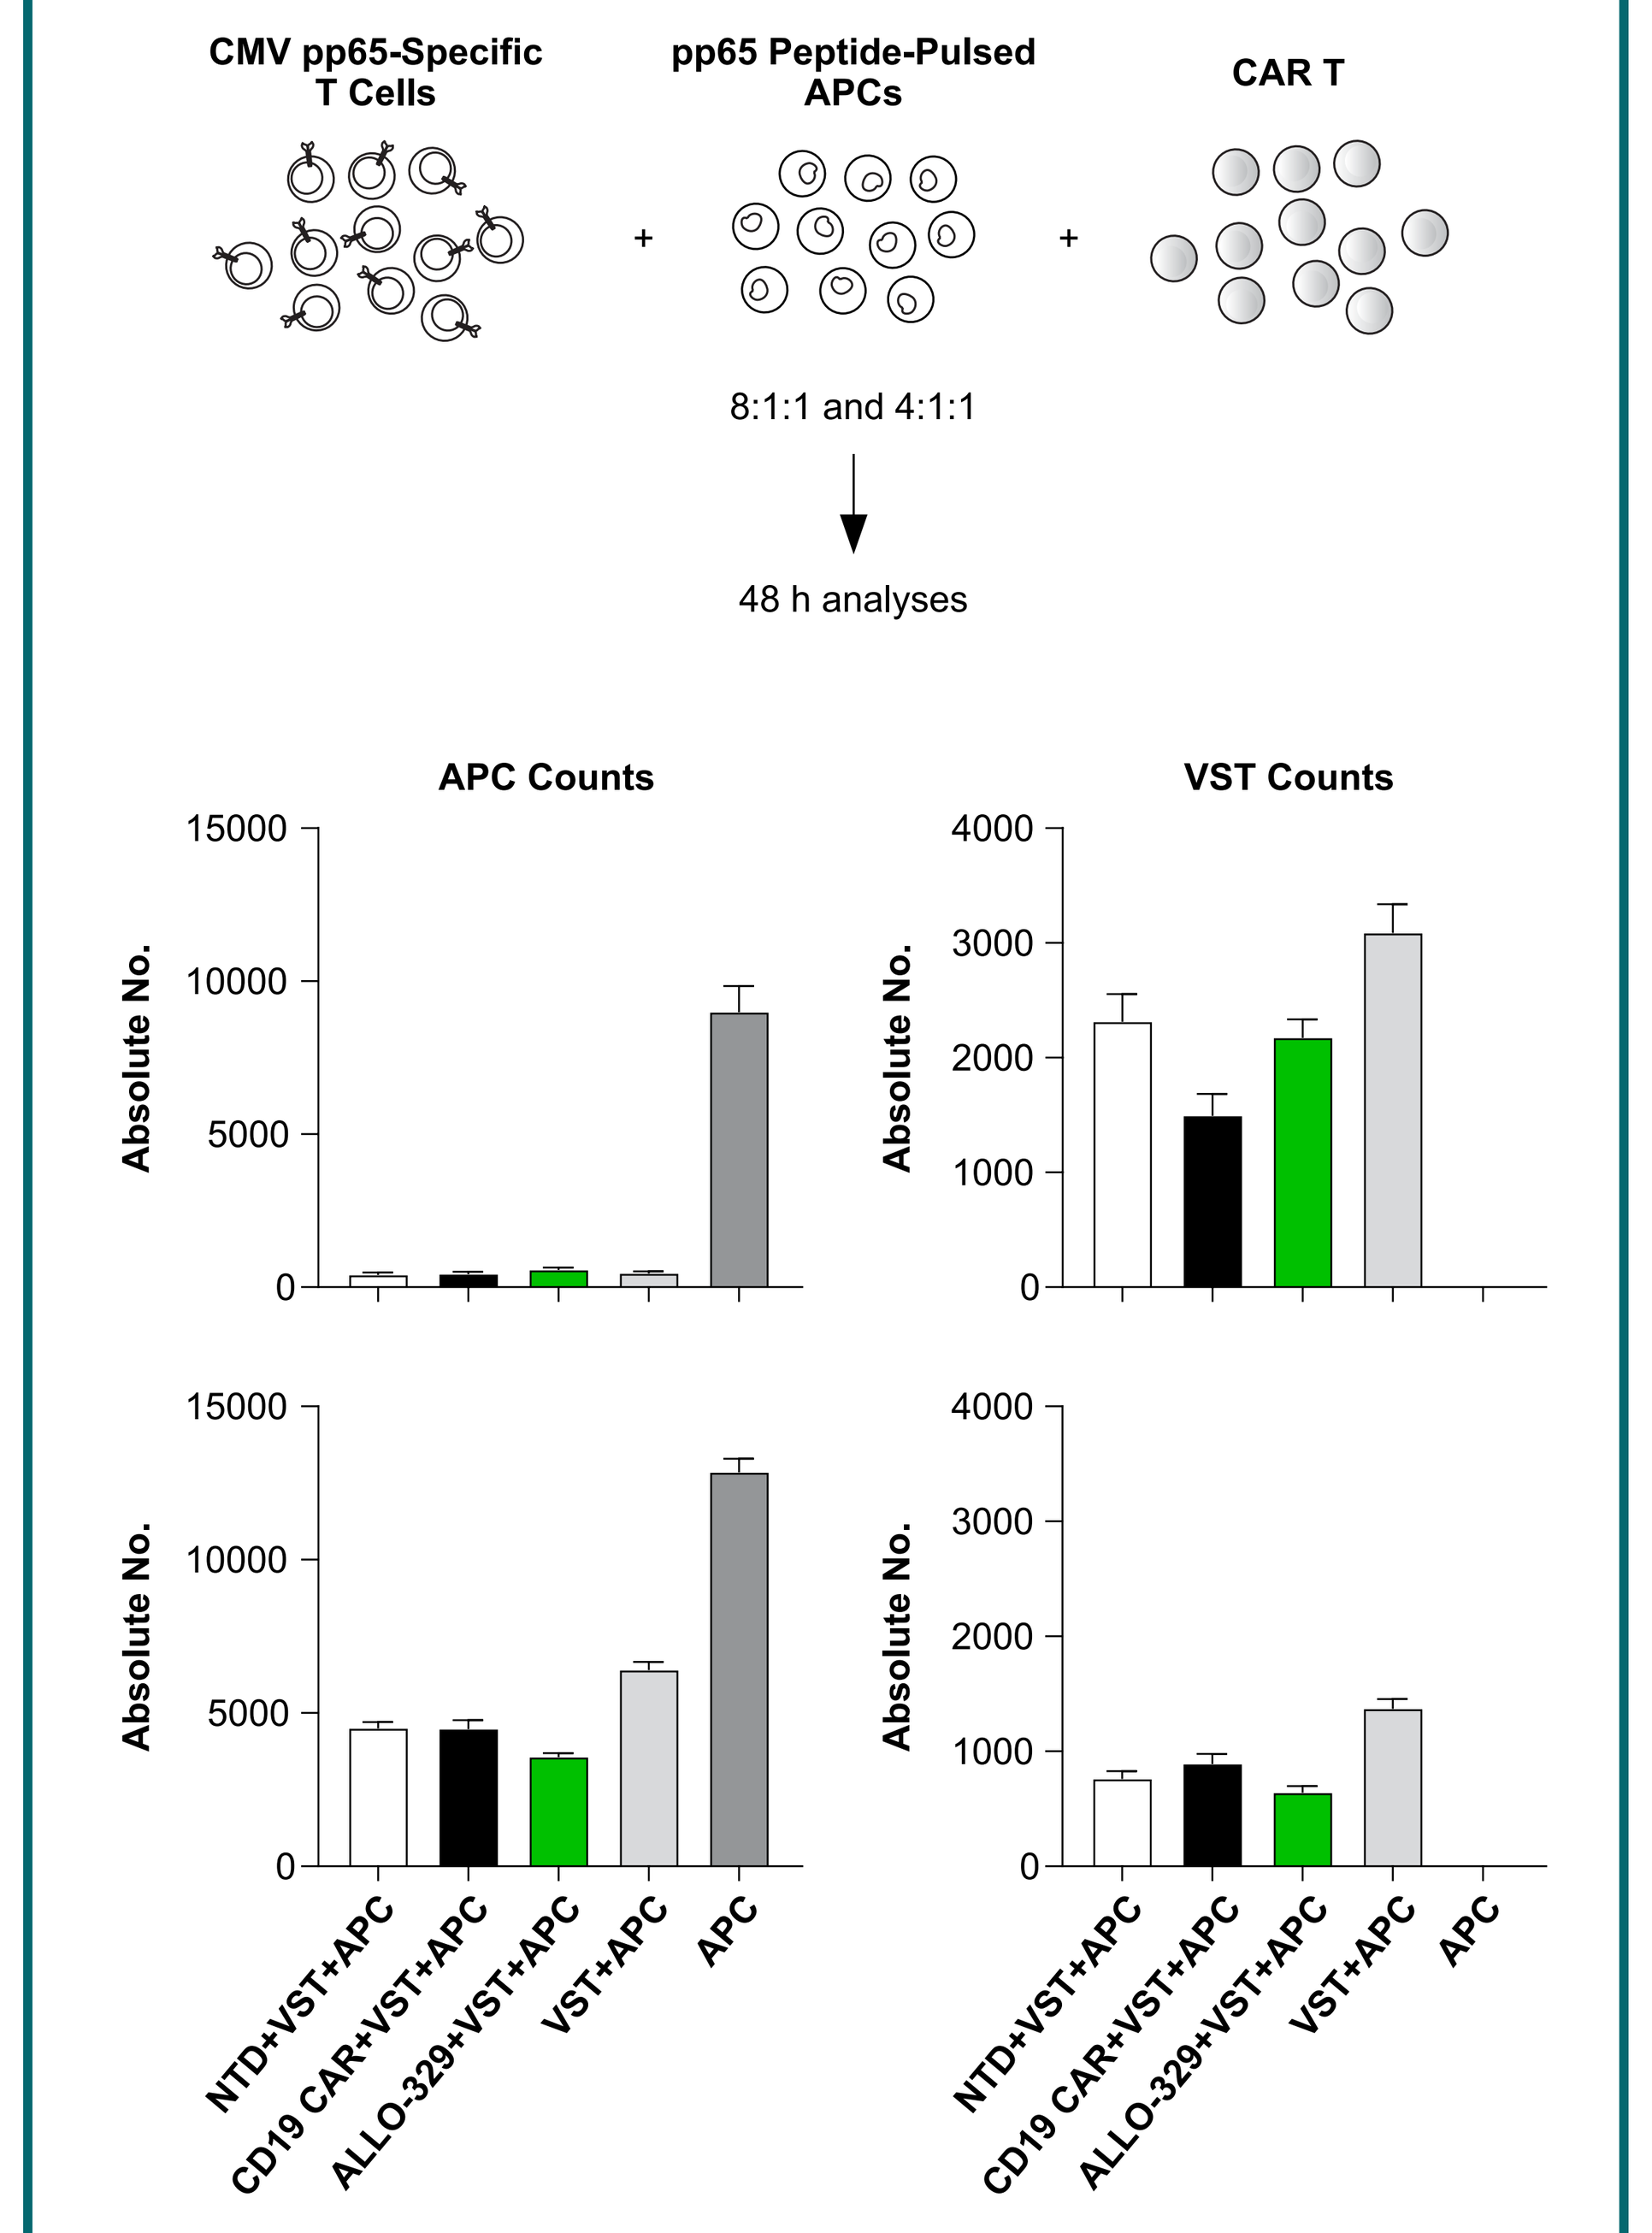
CMV pp65-Specific T Cells
pp65 Peptide-Pulsed APCs
CAR T
8:1:1 and 4:1:1
48 h analyses
APC Counts
VST Counts
Absolute No.
NTD+VST+APC
CD19 CAR+VST+APC
ALLO-329+VST+APC
VST+APC
APC

Average of 3 technical replicates at 48 h are shown. APC was defined as HLA-A2⁺CD3⁻TCR⁻CD20⁺. VST was defined as HLA-A2⁺CD3⁺TCR⁺CD20⁻. CAR T and NTD cells were gated out based on HLA-A2 expression. Results from a single graft:host donor pair. 8:1:1 (top row). 4:1:1 (bottom row).
APC, antigen-presenting cell; CAR, chimeric antigen receptor; CMV, cytomegalovirus; HLA, human leukocyte antigen; KO, knockout; NTD, non-transduced T cells; VST, virus specific T cell.

CONCLUSIONS

- ALLO-329, an allogeneic CD19/CD70 dual CAR T cell, is produced via CRISPR Cas-mediated site-specific integration of a single bicistronic construct in the *TRAC* locus
- ALLO-329 offers (1) a unique ability to deplete both B cells and activated CD70⁺ T cells and (2) the potential to reduce or eliminate the need for concomitant lymphodepletion typically required prior to CAR T cell infusion
 - ALLO-329 exhibited cytolytic activity and target-mediated expansion in the presence of tumor cell lines expressing CD19 and/or CD70
 - Primary CD19⁺ B cells and CD70⁺ T cells from healthy donors and patients with SLE were rapidly eliminated by ALLO-329 *in vivo*, resulting in a concomitant reduction of antibody production
 - ALLO-329 engrafted, expanded, and depleted B cells in hematopoietic stem cell-engrafted mice without the need for irradiation or lymphodepletion
- Plans for clinical evaluation of ALLO-329 in patients with autoimmune disorders are underway

DISCLOSURES

All authors are current employees of Allogene Therapeutics, Inc.

ACKNOWLEDGMENTS

Medical writing and editorial assistance were provided by Second City Science and funded by Allogene Therapeutics, Inc.

Contact Allogene Therapeutics for permission to reprint and/or distribute.

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from ACR and the authors of this poster.

Presented at the American College of Rheumatology Convergence, November 14-19, 2024