Preclinical Evaluation of Allogeneic BCMA/CD70 Dual CAR T Cells for High-Risk Multiple Myeloma

Poster 1535

Mark K. O'Dair, David Qu, Duy Nguyen, Kristen Zhang, Zachary Roberts, Elvin J. Lauron, Cesar Sommer

Allogene Therapeutics, South San Francisco, CA, USA

INTRODUCTION

- Chimeric antigen receptor (CAR) T cells targeting B-cell maturation antigen (BCMA) have demonstrated transformative efficacy in multiple myeloma and are now being actively explored for the treatment of autoimmune diseases[1]
- Off-the-shelf allogeneic CAR T cells manufactured from healthy donor T cells could potentially address some of the limitations of autologous therapies by providing a more consistent product, immediate availability, and the convenience of scalable manufacturing[2]
- However, premature rejection of allogeneic CAR T cells may limit persistence and clinical responses[2]
- To address this challenge, an anti-rejection CD70 CAR was developed to selectively eliminate $CD70^+$ alloreactive T cells and overcome rejection[3]
- Recent studies reported high CD70 expression in tumors from patients with high-risk MM, supporting the evaluation of CD70-targeted therapies in this patient population[4,5]
- Here, we evaluated the ability of allogeneic CAR T cells targeting BCMA and CD70 to overcome rejection and BCMA loss in preclinical models of MM

METHODS

- Human T cells were engineered with CRISPR gene-editing technology to knock out the *TRAC* locus and to allow site-specific integration of a BCMA/CD70 dual CAR construct (**Figure 1**)
- VHH, a CAR containing the BCMA VHH binder from ciltacabtagene autoleucel (cilta-cel), was used as a control in certain experiments
- *TRAC* KO dual CAR T cells were characterized by flow cytometry and through various *in vitro* assays, including cytotoxicity, cytokine release, target-mediated expansion, and mixed lymphocyte reactions
- *In vivo* efficacy and expansion of dual CAR T cells were evaluated in NSG mice bearing tumors following tail vein injection of the human MM cell line MOLP-8
- All data points represent the average ± the standard deviation, unless indicated otherwise
- All procedures performed on animals were reviewed and approved by an Institutional Animal Care and Use Committee and were conducted in accordance with established guidelines
- Additional methodological details are provided in figure legends

RESULTS

Allogeneic BCMA/CD70 Dual CAR T Cells for Multiple Myeloma

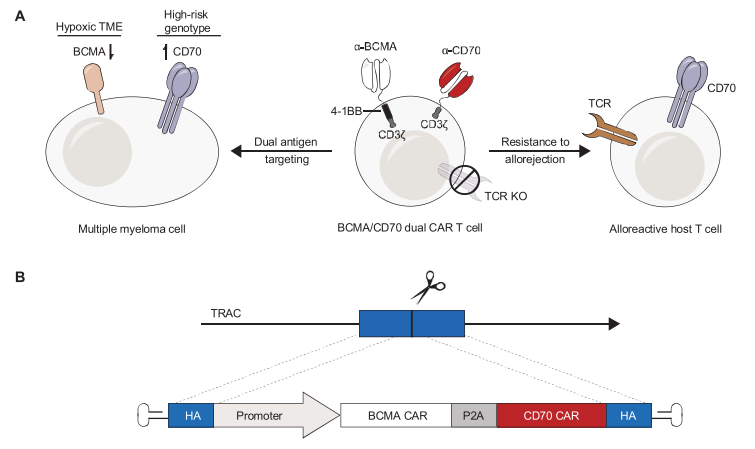
A
Hypoxic TME
BCMA
High-risk genotype
CD70
α-BCMA
α-CD70
4-1BB
CD3ζ
CD3ζ
TCR
CD70
Dual antigen targeting
Resistance to allorejection
TCR KO
Multiple myeloma cell
BCMA/CD70 dual CAR T cell
Alloreactive host T cell
B
TRAC
HA
Promoter
BCMA CAR
P2A
CD70 CAR
HA

Figure 1. (A) Potential benefits of dual targeting of MM with allogeneic BCMA/CD70 dual CAR T cells. Under hypoxic TME conditions, MM cells downregulate BCMA surface expression, whereas patients with certain high-risk genotypes show upregulation of CD70.[4,6] BCMA/CD70 dual CAR expression enables simultaneous targeting of MM cells and $CD70^+$ alloreactive host T cells, providing enhanced targeting of MM with potential to overcome BCMA antigen loss and resistance to allorejection. (B) The dual CAR transgene is integrated into the *TRAC* locus. The BCMA CAR contains an scFv derived from a fully human anti-BCMA antibody, a hinge and transmembrane domain, and intracellular domains of 4-1BB (costimulatory domain) and CD3ζ (activation domain), whereas the CD70 CAR contains an anti-CD70 scFv, a hinge and transmembrane domain, and the CD3ζ domain only without a costimulatory domain.

BCMA/CD70 Dual CAR T Cells Exhibit Similar Expansion, Activation, and Phenotype Compared With BCMA VHH/CD70 Dual CAR T Cells

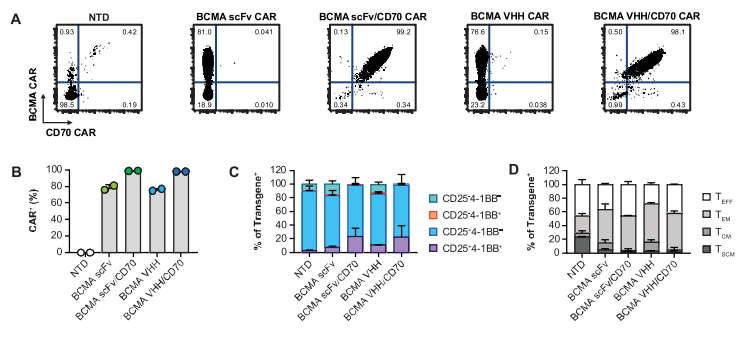
A
NTD
BCMA scFv CAR
BCMA scFv/CD70 CAR
BCMA VHH CAR
BCMA VHH/CD70 CAR
BCMA CAR
CD70 CAR
B
CAR+ (%)
C
% of Transgene+
D
NTD
BCMA scFv
BCMA scFv/CD70
BCMA VHH
BCMA VHH/CD70

Figure 2. (A) Representative flow cytometry plots showing BCMA CAR and CD70 CAR expression on human T cells (Day 15). Stained with biotinylated soluble BCMA and anti-idiotype antibody against the CD70 CAR. (B) Percentage of CAR^+ (single or dual) on TCR^- T cell fraction at the end of production. (C) Expression of activation markers CD25 and 4-1BB on CAR^+ cells midway through production (Day 9). (D) Distribution of memory and effector CAR T cell subsets as determined by the surface expression of CCR7 and CD45RA (Day 15). Data are presented as the percentage of effector, effector memory, central memory, and stem cell memory CAR^+ cells.

BCMA/CD70 Dual CAR T Cells Exhibit Sustained *In Vitro* and *In Vivo* Activity Similar to CAR T Cells With the VHH Binder

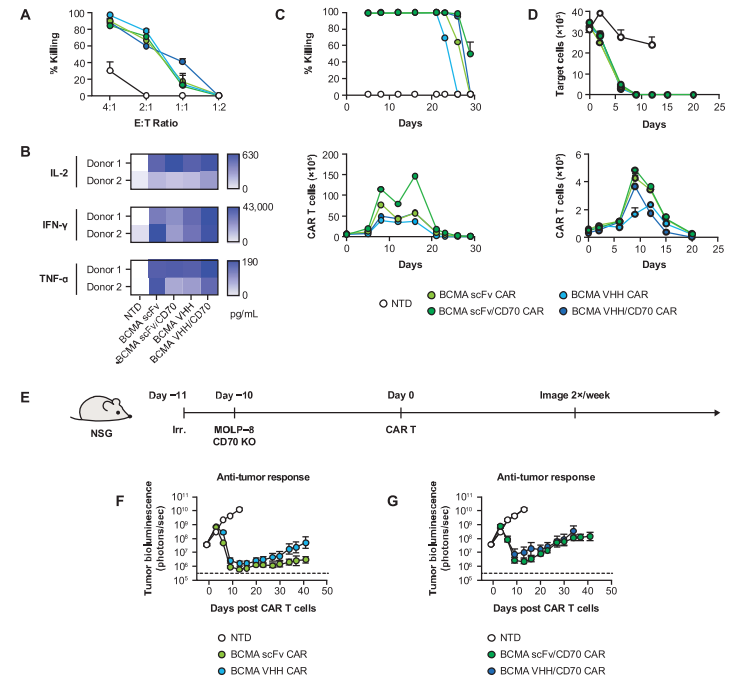
A
% Killing
E:T Ratio
C
Days
D
Target cells (×10⁶)
B
IL-2
IFN-γ
TNF-α
Donor 1
Donor 2
pg/mL
CAR T cells (×10⁶)
NTD
BCMA scFv CAR
BCMA scFv/CD70 CAR
BCMA VHH CAR
BCMA VHH/CD70 CAR
E
NSG
Day −11
Day −10
Day 0
Image 2×/week
Irr.
MOLP–8 CD70 KO
CAR T
F
G
Anti-tumor response
Tumor bioluminescence (photons/sec)
Days post CAR T cells

Figure 3. (A) Overnight killing assay at varying effector to target cell ratios. (B) Cytokine secretion from CAR T cells following stimulation with target cells. (C) Serial restimulation assay. Effector cells were co-cultured with target cells at a ratio of 1:3. At each timepoint, new target cells were added. Killing of target cells (top) and CAR T cell expansion (bottom). (D) High tumor burden proliferation assay. Effector cells were stimulated with target cells at a ratio of 1:30. Target cell counts (top) and CAR T cell expansion (bottom). (E) MOLP-8 CD70 KO cells (2×10^6) were injected into NSG mice 10 days before CAR T cell treatment ($n = 10$, 3×10^6 dose intravenously). (F) and (G) Anti-tumor activity of single (left) and dual (right) CAR T cells determined by bioluminescence imaging. Data represent results from 2 unique donors. *In vivo* data are given as mean ± standard error of the mean. All *in vitro* assays employed MOLP-8 CD70 KO as target cells.

CD70 CAR Protects BCMA CAR T Cells Against Allorejection

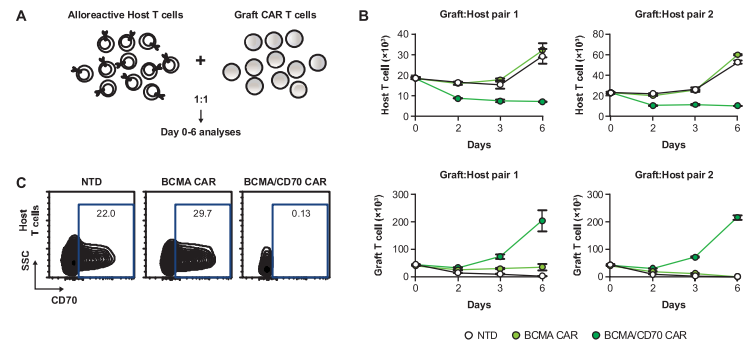
A
Alloreactive Host T cells
Graft CAR T cells
1:1
Day 0-6 analyses
B
Graft:Host pair 1
Graft:Host pair 2
Host T cell (×10³)
Graft T cell (×10³)
Days
C
NTD
BCMA CAR
BCMA/CD70 CAR
Host T cells
SSC
CD70
22.0
29.7
0.13
NTD
BCMA CAR
BCMA/CD70 CAR

Figure 4. (A) Schematic of the primed T cell MLR assay. Alloreactive host T cells were produced by priming T cells from allogeneic donors against unedited graft T cells. Host T cells were then mixed with graft CAR T cells. (B) Cell counts of host T cells (top) and graft T cells (bottom) for 2 different alloreactive host donors (left and right). Data are representative of a total of 6 unique graft:host pairs. (C) Representative flow cytometry plots of CD70 expression on host T cells on Day 6.

Allogeneic BCMA/CD70 Dual CAR T Cells Avoid Allorejection and Deplete Plasma Cells in a Humanized Mouse Model

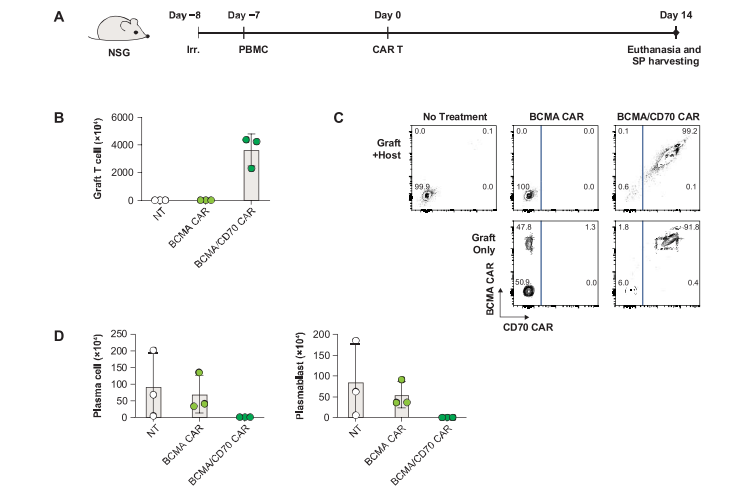
A
NSG
Day −8
Day −7
Day 0
Day 14
Irr.
PBMC
CAR T
Euthanasia and SP harvesting
B
Graft T cell (×10³)
NT
BCMA CAR
BCMA/CD70 CAR
C
No Treatment
BCMA CAR
BCMA/CD70 CAR
Graft +Host
Graft Only
BCMA CAR
CD70 CAR
D
Plasma cell (×10³)
Plasmablast (×10³)

Figure 5. (A) Schematic of PBMC *in vivo* rejection model. (B) Cell counts of graft CAR T cells in the spleen. (C) Representative flow cytometry plots of graft BCMA CAR and BCMA/CD70 dual CAR T cells in the spleen. (D) Cell counts of plasma cells (left) and plasmablast cells (right) in the spleen. Symbols represent individual mice ($n = 3$). All data shown are from the spleen on Day 14. Data are representative of 2 PBMC donors tested.

BCMA/CD70 Dual CAR T Cells Show High Potency Across Multiple Donors in an Aggressive MOLP-8 *In Vivo* Model

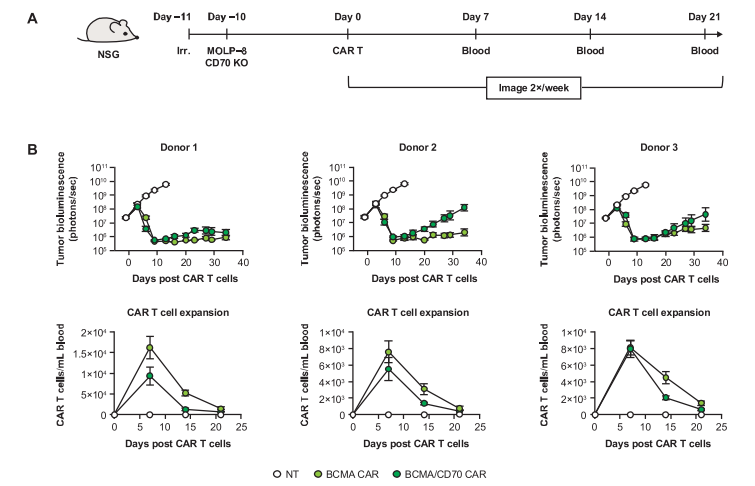
A
NSG
Day −11
Day −10
Day 0
Day 7
Day 14
Day 21
Irr.
MOLP–8 CD70 KO
CAR T
Blood
Blood
Blood
Image 2×/week
B
Donor 1
Donor 2
Donor 3
Tumor bioluminescence (photons/sec)
Days post CAR T cells
CAR T cell expansion
CAR T cells/mL blood
NT
BCMA CAR
BCMA/CD70 CAR

Figure 6. Efficacy and expansion of single and dual CAR T cells in NSG mice bearing MOLP-8 tumor xenografts. (A) Study design. MOLP-8 CD70 KO cells (2×10^6) were injected into NSG mice 10 days before CAR T cell treatment ($n = 8$, 3×10^6 dose intravenously). Tumors were monitored using bioluminescence imaging, and blood samples were collected to assess CAR T cell expansion. (B) Anti-tumor activity (top) and CAR T cell expansion in the peripheral blood (bottom). *In vivo* data are given as mean ± standard of the mean.

BCMA/CD70 Dual CAR T Cells Eliminate Antigenically Heterogeneous Tumor Cell Populations

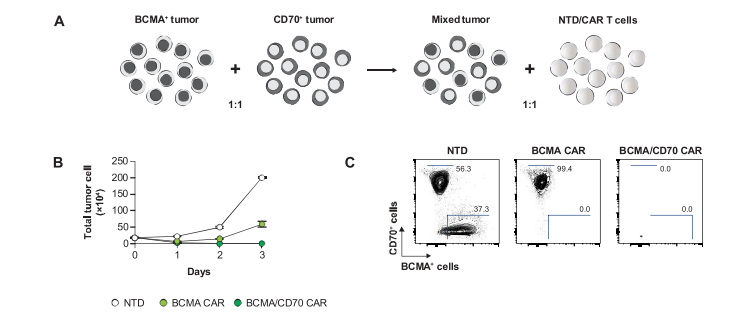
A
BCMA⁺ tumor
CD70⁺ tumor
Mixed tumor
NTD/CAR T cells
1:1
1:1
B
Total tumor cell (×10³)
Days
C
NTD
BCMA CAR
BCMA/CD70 CAR
CD70⁺ cells
BCMA⁺ cells
NTD
BCMA CAR
BCMA/CD70 CAR

Figure 7. (A) Schematic of mixed tumor assay. K562 tumor cells overexpressing either BCMA or CD70 antigen were mixed at a 1:1 ratio then cultured with NTD or CAR T cells. At each timepoint, samples were taken and analyzed by flow cytometry. (B) Total tumor cell counts of mixed $BCMA^+$ and $CD70^+$ K562 cells. (C) Representative flow cytometry plots of K562 tumor cell mixture on day 4 of culture with NTD or CAR T cells. Data are representative of two CAR T cell donors tested.

CONCLUSIONS

- BCMA/CD70 dual CAR T cells produced using site-specific integration show specific cytotoxic activity, rejection avoidance, and the potential to eliminate tumor cells that have downregulated BCMA
- Our findings underscore the promise of allogeneic BCMA/CD70 dual CAR T cells as an accessible, off-the-shelf treatment option for patients with high-risk MM with potential to extend toward broader applications in autoimmune disorders

REFERENCES

1. Yu B, et al. *J Hematol Oncol.* 2020;13(1):125.
2. Depil S, et al. *Nat Rev Drug Discov.* 2020;19:185-199.
3. Zhang K, et al. *Arthritis Rheumatol.* 2024;76(suppl 9):1841.
4. Kasap C, et al. *Blood.* 2025;146:819-833.
5. Lin P, et al. *Blood Cancer Discov.* 2026;7:234-239.
6. Tu C, et al. *Exp Hematol Oncol.* 2026;15:1.

ABBREVIATIONS

BCMA, B-cell maturation antigen; CAR, chimeric antigen receptor; CD3ζ, cluster of differentiation 3 zeta; CD70, cluster of differentiation 70; Cilta-cel, ciltacabtagene autoleucel; CRISPR, Clustered Regularly Interspaced Short Palindromic Repeats; E:T, effect to target cell ratio; HA, homology arm; IFN, interferon; Irr, irradiate; KO, knock out; MLR, mixed lymphocyte reaction; MM, multiple myeloma; NSG, NOD-scid IL2Rγnull; NT, not treated; NTD, non-transduced TCR-negative cell; P2A, self-cleaving 2A peptide; PBMC, peripheral blood mononuclear cell; scFv, single-chain variable fragment; SP, spleen; SSC, side scatter; T_{CM}, central memory cell; TCR, T cell receptor; T_{EFF}, effector cell; T_{EM} effector memory cell; TME, tumor microenvironment; TNF, tumor necrosis factor; TRAC, T cell receptor α constant gene; TSCM, stem cell memory cell; VHH, variable domain of heavy-chain-only antibody.

ACKNOWLEDGMENTS

The authors thank Zhe Li and Adam Mealy (Allogene Therapeutics, Inc) for their assistance in designing and running *in vivo* studies, and Lynn Brown, PhD (ApotheCom, Yardley, PA), for editorial and writing assistance, which was funded by Allogene Therapeutics, South San Francisco, CA, USA

DISCLOSURES

All authors are current or former employees of Allogene Therapeutics, Inc

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at the AACR Annual Meeting; April 17-22, 2026; San Diego, CA, USA